A Novel Recombinant Human Acid Alpha-Glucosidase, ATB200, Leads to Greater Substrate Reduction and Improvement in Pompe Disease-Relevant Markers Compared to Alglucosidase Alfa in *Gaa* KO Mice

Lun Y, Xu S, Soska, R, Nair A, Frascella M, Garcia A, Gotschall R, Do H, Valenzano KJ, and Khanna R
Amicus Therapeutics, 1 Cedar Brook Drive, Cranbury, NJ 08512, USA

Introduction

Pompe disease is an inherited lysosomal storage disorder caused by a deficiency in acid alpha-glucosidase (GAA) activity, and is characterized by progressive accumulation of lysosomal glycogen in cardiac and skeletal muscles. Enzyme replacement therapy (ERT) using recombinant human GAA (rhGAA), alglucosidase alfa, is the only approved treatment available for Pompe disease. While alglucosidase alfa provides some clinical benefits, the infused enzyme shows insufficient uptake into key disease-relevant skeletal muscles.[1] This distribution is likely due to sub-optimal levels of mannose-6-phosphate (M6P), a carbohydrate that binds cation-independent M6P receptors (CI-MPR) at the cell surface resulting in enzyme internalization and lysosomal targeting. In order to increase the targeting efficiency of ERT, we have developed a novel rhGAA (designated ATB200) with a significantly higher M6P content compared to alglucosidase alfa. The pharmacological chaperone AT2221 is also utilized to stabilize ATB200 in blood. In this study, we compared the effects of repeat co-administration of ATB200 and AT2221 (ATB200 / AT2221) to those of alglucosidase alfa in *Gaa* KO mice.

1. ATB200 Has a Higher M6P Content Compared with Alglucosidase Alfa

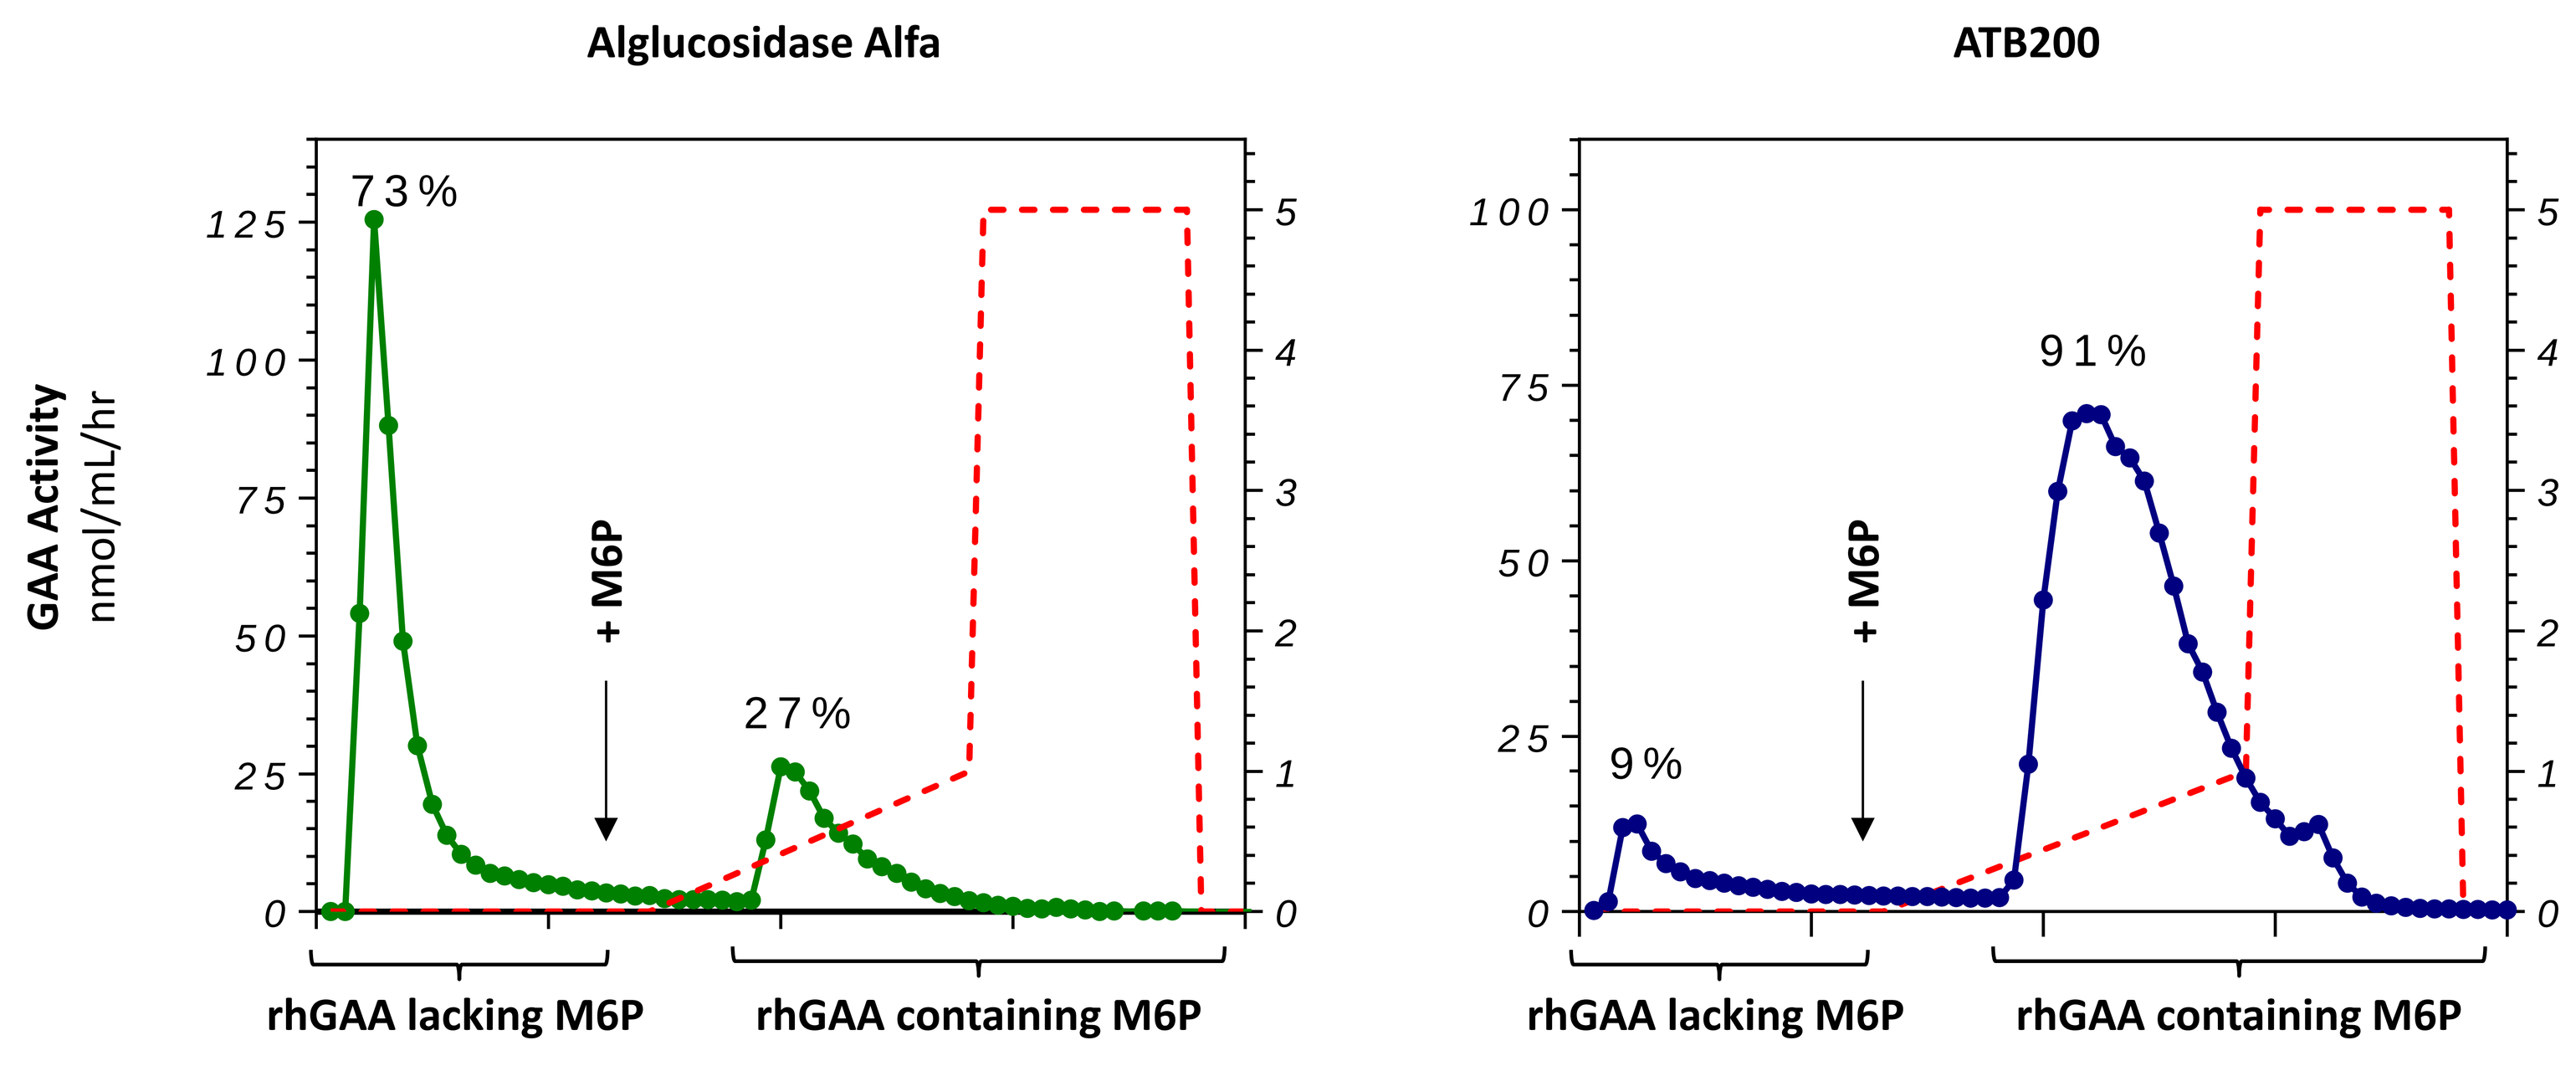
Alglucosidase Alfa
ATB200
GAA Activity
nmol/mL/hr
73%
27%
9%
91%
+ M6P
rhGAA lacking M6P
rhGAA containing M6P

The M6P content was measured by affinity chromatography, where alglucosidase alfa or ATB200 was loaded onto a CI-MPR column. Only enzyme that contained M6P was retained and then eluted from the column using free M6P of increasing concentration (dotted line in red). Both M6P-lacking and M6P-containing rhGAA fractions were collected and assayed for GAA activity. The majority of ATB200 (91%) was bound compared to alglucosidase alfa (27%), suggesting that ATB200 has a higher M6P content, which is key to the efficient endocytosis and lysosomal targeting of rhGAA.

2. Pharmacological Chaperone AT2221 Stabilizes ATB200 at Neutral pH *in vitro*

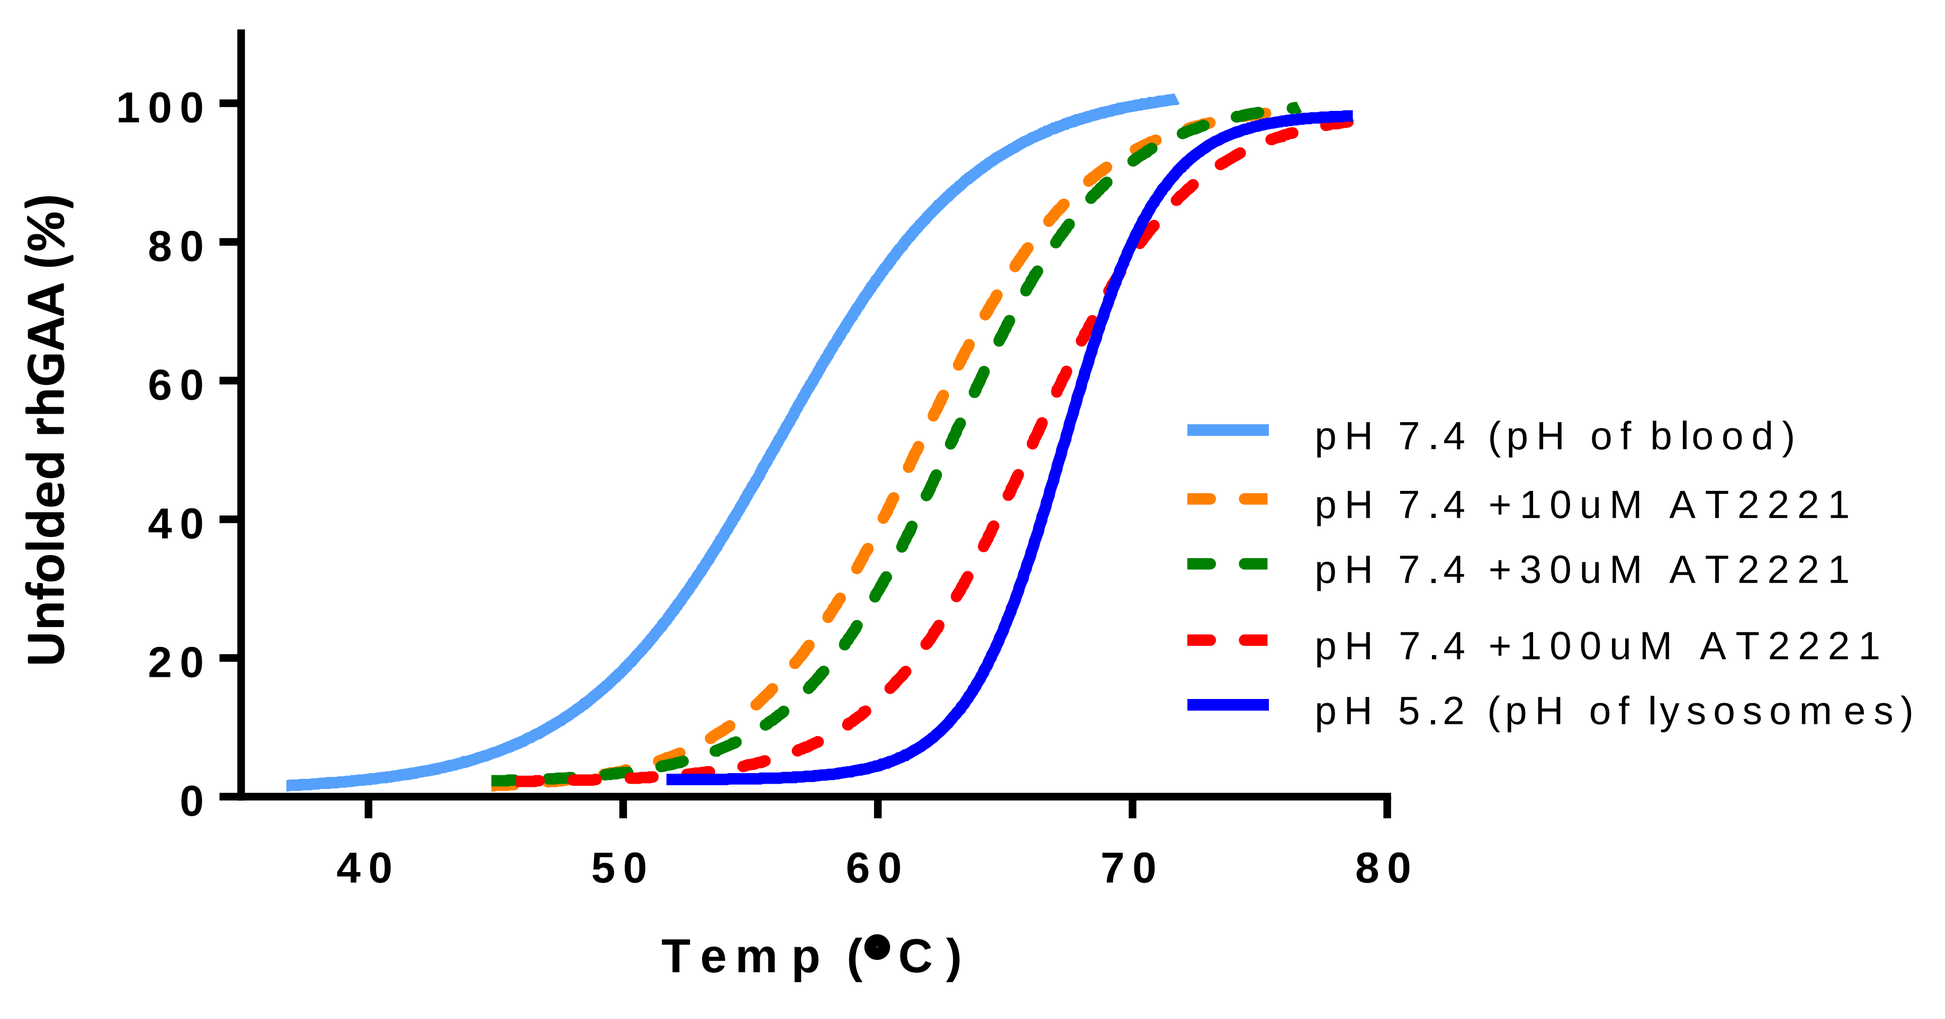
Unfolded rhGAA (%)
Temp (°C)
pH 7.4 (pH of blood)
pH 7.4 +10uM AT2221
pH 7.4 +30uM AT2221
pH 7.4 +100uM AT2221
pH 5.2 (pH of lysosomes)

Test Condition	Tm (°C)
pH 7.4	56.2
pH 7.4 + 10 μm AT2221	61.6
pH 7.4 + 30 μm AT2221	62.9
pH 7.4 + 100 μm AT2221	66.0
pH 5.2	67.3

The stability of ATB200 in acidic or neutral pH buffers was evaluated in a thermostability assay using SYPRO Orange, as the fluorescence of the dye increases when proteins denature. AT2221 stabilizes ATB200 at pH 7.4 in a concentration-dependent manner, to that approaching the level seen at pH 5.2, a condition that mimics the acidic environment of the lysosome, as demonstrated by a nearly 10°C increase in the melting temperature (T_m) of ATB200. Thus, we hypothesized that ATB200 may also be stabilized by AT2221 in circulation.

3. Study Design: Co-administration of ATB200 and AT2221 in *Gaa* KO Mice

Genotype	Treatment	Drug Dosage per Administration (bi-weekly)	Number of Administration
Gaa KO	Vehicle	Not applicable	6
Gaa KO	Alglucosidase Alfa	20 mg/kg	6
Gaa KO	ATB200 / AT2221	20 mg/kg / 10 mg/kg	6
WT (Sve 129)	Untreated	Not applicable	Not applicable

- Male *Gaa* KO (3- to 4-month old) and age-matched wild-type (WT) mice were used in the study.
- Alglucosidase alfa was administered via bolus tail vein intravenous (IV) injection.
- In the co-administration regimen, AT2221 was administered via oral gavage (PO) 30 minutes prior to the IV injection of ATB200.
- Tissues were collected 14 days following the last administration.

Reference

1. Geel TM et al. *Mol Genet Metab*. 2007; 92: 299-307

4. ATB200 / AT2221 Led to Greater Glycogen Reduction Compared to Alglucosidase Alfa in Muscles of *Gaa* KO Mice

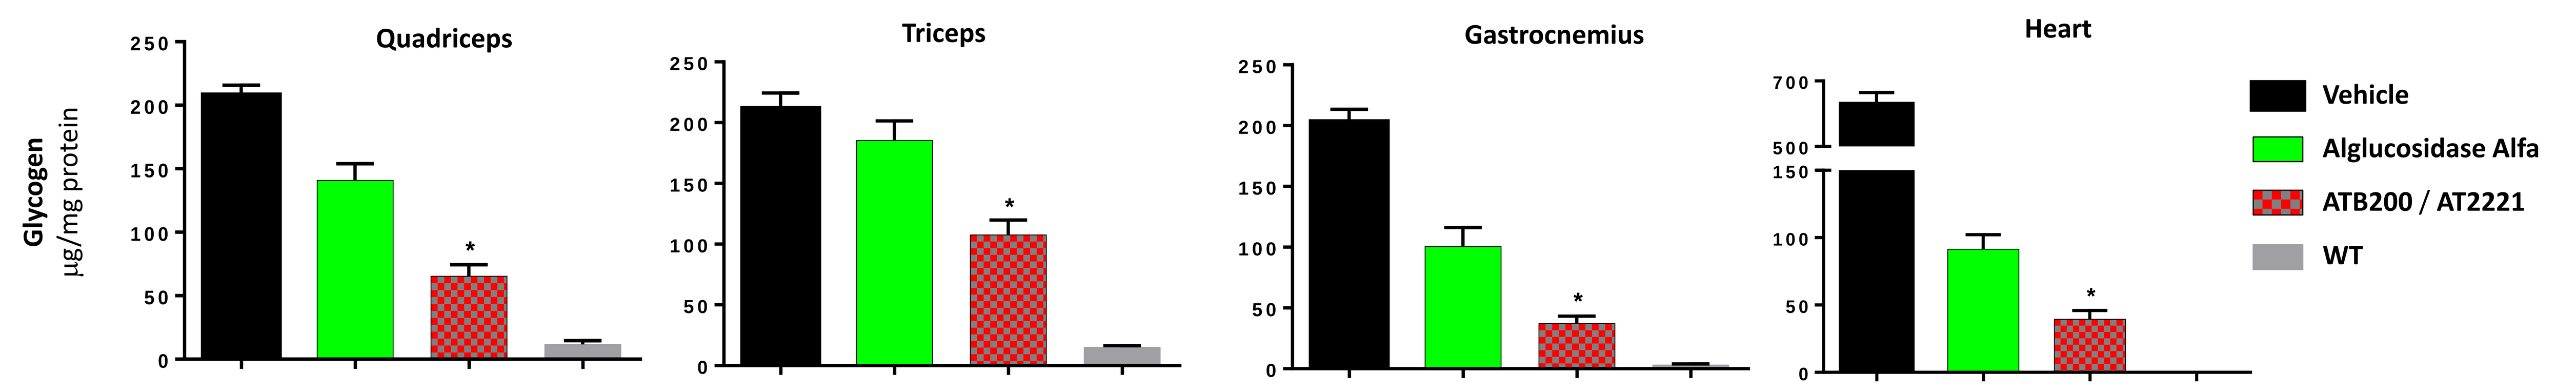
Glycogen
μg/mg protein
Quadriceps
Triceps
Gastrocnemius
Heart
Vehicle
Alglucosidase Alfa
ATB200 / AT2221
WT

Tissue glycogen content was determined using amyloglucosidase digestion. Bars represent Mean ± SEM of 7 mice/group. * p<0.05 compared to alglucosidase alfa in multiple comparison using Dunnett's method under one-way ANOVA analysis.

5. ATB200 / AT2221 Reduced LAMP1-Positive Vesicles More Effectively than Alglucosidase Alfa in Quadriceps of *Gaa* KO Mice

IHC Examination of LAMP1 in *vastus lateralis* of Quadriceps

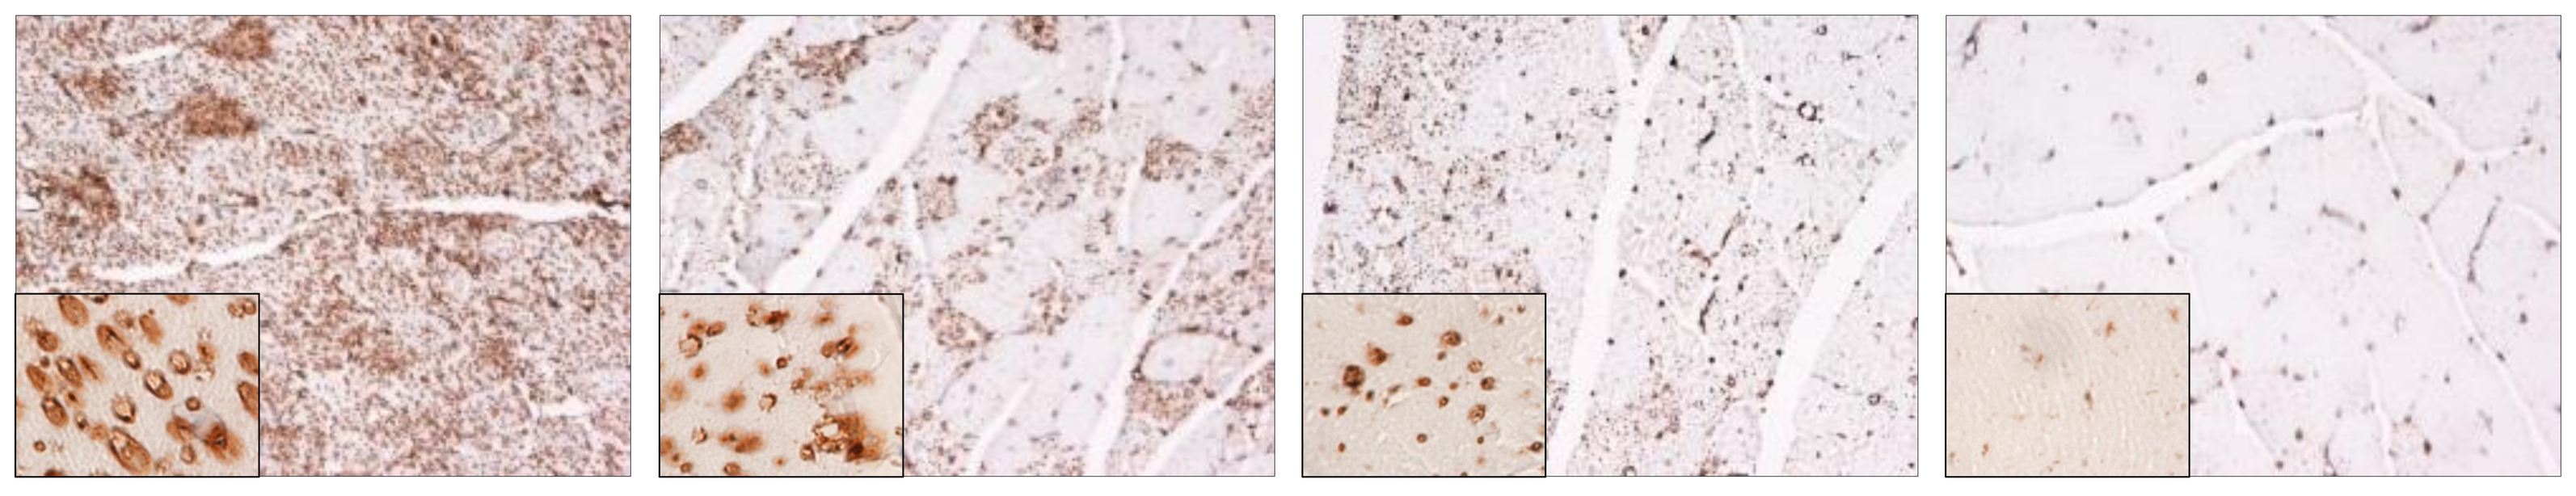

Vehicle | Alglucosidase Alfa | ATB200 / AT2221 | WT

A profound increase in and enlargement of LAMP1-positive vesicles was seen in muscle fibers of *Gaa* KO animals compared to WT, indicative of lysosomal proliferation. Co-administration of ATB200 / AT2221 led to more fibers with normalized LAMP1 level, while the remaining LAMP1-positive vesicles also reduced in size (insets). Images were taken from *vastus lateralis* and were representative of 7 mice per group. Magnification = 200x (1,000x in insets).

6. ATB200 / AT2221 Reduced LC3 II Levels More Effectively than Alglucosidase Alfa in Quadriceps of *Gaa* KO Mice

A. IHC Examination of LC3 in *vastus lateralis* of Quadriceps

B. WB Examination of LC3 in Quadriceps

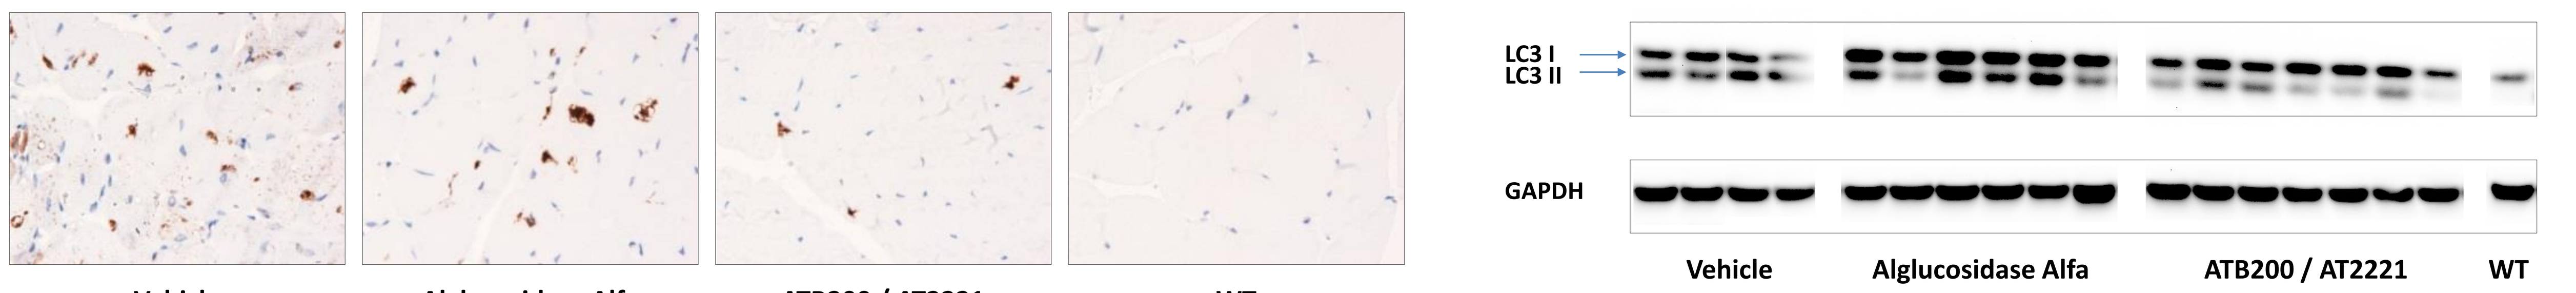
LC3 I
LC3 II
GAPDH
Vehicle
Alglucosidase Alfa
ATB200 / AT2221
WT

A. Intense LC3-positive aggregates in the muscle fibers of untreated *Gaa* KO mice signify the presence of autophagic zones and autophagy build-up. They were preferentially reduced with ATB200 / AT2221 co-administration compared to alglucosidase alfa. Images were taken from *vastus lateralis* and were representative of 7 mice per group. Magnification = 400x.
B. The majority of animals treated with ATB200 / AT2221 showed a significant decrease in levels of LC3 II, the lipidated form that is associated with autophagosomes, suggesting an improved autophagy flux. In comparison, the effect of alglucosidase alfa on autophagy was modest. A total of 30 μg protein was loaded in each lane.

7. ATB200 / AT2221 Corrected Dysferlin Mistrafficking More Effectively than Alglucosidase Alfa in Quadriceps of *Gaa* KO Mice

IHC Examination of Dysferlin in *vastus lateralis* of Quadriceps

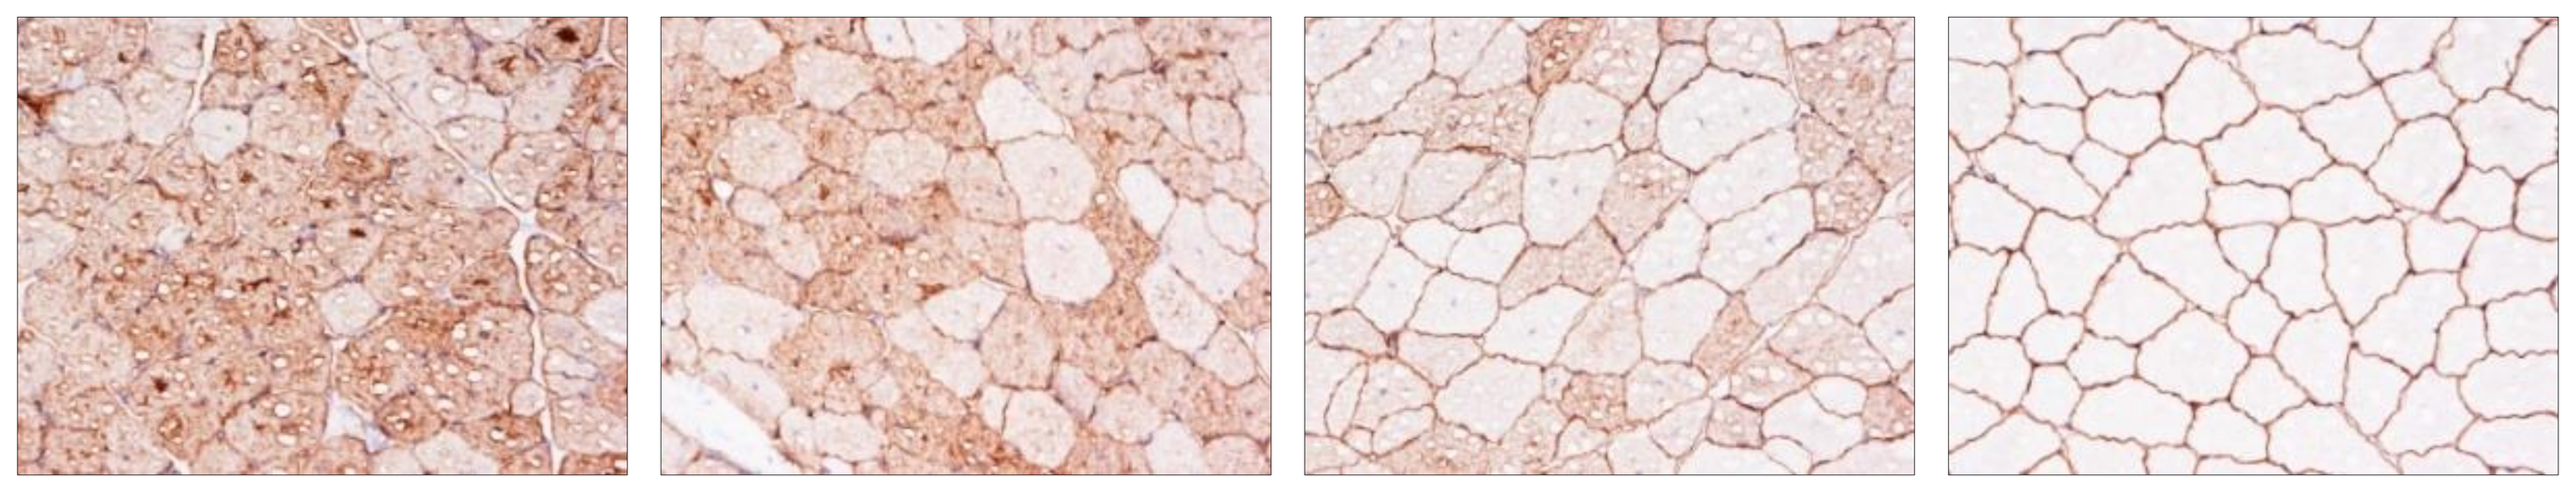

Vehicle | Alglucosidase Alfa | ATB200 / AT2221 | WT

Dysferlin, a protein involved in membrane repair and whose deficiency/mistrafficking is associated with a number of muscular dystrophies, was heavily accumulated in the sarcoplasm of *Gaa* KO mice. Compared to alglucosidase alfa, ATB200 / AT2221 was able to restore dysferlin to the sarcolemma in a greater number of muscle fibers. Images were taken from *vastus lateralis* and were representative of 7 mice per group. Magnification = 200x.

Summary and Conclusions

- Our data suggest that ATB200, with its higher M6P content and further stabilization by the pharmacological chaperone AT2221 at the neutral pH of blood, is more efficient in tissue targeting and lysosomal trafficking compared to alglucosidase alfa when administered to *Gaa* KO mice.
- As a result, co-administration of ATB200 / AT2221 is more effective than alglucosidase alfa in correcting some of the disease-relevant pathologies, such as glycogen accumulation, lysosomal proliferation, and formation of autophagic zones. In addition, it is likely that ATB200 / AT2221 co-administration may improve the chance of muscle fiber recovery from damage.
- Taken together, co-administration of ATB200 / AT2221 may potentially lead to improved muscle function and thus warrants further investigation as a next-generation treatment for Pompe disease.

Presented at 13th Annual WORLD*Symposium*™; February 13-17, 2017; San Diego, CA, USA

Amicus Therapeutics