Preliminary Results from an Ongoing Phase 2a Study to Investigate the Effect of a Single Dose of Migalastat HCl, a Pharmacological Chaperone, on Agalsidase Activity in Subjects with Fabry Disease

F. Johnson[1], S. Sitaraman[1], M. Adera[1], P. N. Mudd Jr.[2], J. Flanagan[1], R. Khanna[1], K. J. Valenzano[1], D. J. Lockhart[1], and P. Boudes[1] on Behalf of Study AT1001-013 Principal Investigators

[1]Amicus Therapeutics, 6 Cedar Brook Drive, Cranbury, NJ 08512 USA and [2]Glaxo Smith-Kline, 5 Moore Drive, Research Triangle Park, NC 27709 USA

Introduction

Fabry disease is an X-linked lysosomal storage disease caused by mutations in *GLA*, the gene encoding α-Galactosidase A (α-Gal A). Migalastat HCl (AT1001; GR181413A) is a pharmacological chaperone that binds and stabilizes mutant forms of α-Gal A. When co-administered with an Enzyme Replacement Therapy (ERT), such as agalsidase alpha or beta, migalastat HCl is intended to bind to and stabilize the enzyme in the circulation. In preclinical studies, when compared to infusion of agalsidase alone, co-administration of oral migalastat HCl resulted in dose-dependent increases in skin, heart, kidney, and plasma α-Gal A activity and reductions in the substrate of α-Gal A, globotriaosylceramide (GL-3). AT1001-013 (NCT # 01196871) is an ongoing Phase 2a drug-drug interaction study to evaluate the safety and pharmacokinetic (PK) effects of the co-administration of oral migalastat HCl with intravenous agalsidase in male patients with Fabry disease.

Objectives

To evaluate the safety and PK of two doses of migalastat HCl (150 mg and 450 mg) co-administered with ERT (agalsidase) in males diagnosed with Fabry disease.

Study Design and Methods

AT1001-013 is an ongoing, open-label, non-randomized, 2-stage, fixed-sequence study.

- <u>Stage 1</u> is comprised of 3 periods.
 - <u>Period 1</u>: IV infusion of ERT alone.
 - <u>Period 2</u>: migalastat HCl (150 mg) orally administered 2 hours prior to IV infusion of the ERT (at the same dose as in period 1).
 - <u>Period 3</u>: oral administration of migalastat HCl 150 mg alone

Preliminary results are available for Stage 1, Periods 1 and 2:

- Subjects receive their current dose and regimen of agalsidase beta alone at one infusion (0.5 or 1.0 mg/kg for ~2 hrs) followed by oral migalastat HCl 150 mg administered two hours prior to agalsidase beta at their next infusion.
- Due to the supply shortage of agalsidase beta, five of the current seven subjects received 0.5 mg/kg every two weeks and two of the seven subjects received a dose of 1.0 mg/kg every four weeks.
- In stage 2, a 450 mg dose of migalastat HCl will be studied.
- Stages 1 and 2 will be repeated in unique subjects with an ERT infusion of agalsidase alpha (0.2 mg/kg for ~40 min)

<u>Eligible patients</u>
- Male, 18 to 65 years old with Fabry Disease

<u>Key inclusion criteria</u>
- Body Mass Index (BMI) between 18-35;
- Initiated treatment with agalsidase at least 1 month before dosing;
- Estimated creatinine clearance (CLcr) ≥ 50 mL/min at screening.

<u>Key exclusion criteria</u>
- A documented transient ischemic attack, ischemic stroke, unstable angina, or myocardial infarction within 3 months before screening;
- Clinically significant unstable cardiac disease;
- Sensitivity to or concomitant therapy with iminosugars (e.g., miglustat, miglitol)

All subjects gave written informed consent.

Serial blood samples are taken to 24 hours post dose for plasma α-Gal A activity and protein levels each period. Blood samples for α-Gal A activity in peripheral blood mononuclear cells (PBMCs) are taken at predose and on Days 1, 2, 7, and 14 each period. Punch biopsies for skin α-Gal A activity are taken at predose Period 1 and on Days 2 and 7 during Periods 1 and 2. Plasma α-Gal A activity PK parameters includes C_{max}, T_{max}, $AUC_{0\text{-}t}$, $AUC_{0\text{-}inf}$, and t½. Pharmacokinetic parameters are calculated using standard non-compartmental procedures (WINNONLIN version 5.0 or higher).

α-Gal A activity in plasma, skin, and PBMC lysates are measured by a fluorescence enzyme assay using 4-methylumbelliferyl-α-D-galactopyranoside (4-MUG). α-Gal A activity on 4-MUG is measured in vitro following serial dilutions to dissociate migalastat.

Western blot analysis of α-Gal A protein are performed on plasma samples using anti-human α-Gal A antibody. An rha-Gal A (agalsidase) standard curve was run to calculate the appropriate concentration of α-Gal A protein in each sample.

The safety parameters includes adverse events (AEs), vital signs, clinical laboratory tests (hematology, serum chemistry, and urinalysis), electrocardiograms (ECGs), physical examinations, and use of concomitant medications.

Acknowledgements

The Principal Investigators for AT1001-013 currently include Drs. David Warnock (University of Alabama at Birmingham School of Medicine, Division of Nephrology), Ozlem Goker-Alpan (O & O Alpan LLC), Suma Shankar (Emory University School of Medicine, Emory Genetics Clinic), Myrl Holida (University of Iowa Children's Hospital), and Gabor Linthorst (Academisch Medisch Centrum, Endocrinology and Metabolism Department).

Preliminary Results

Patient Disposition and Demographics

Seven patients with plasma, skin and PBMC α-Gal A activity from Stage 1, Periods 1 and 2 were evaluated.

- All 7 patients received agalsidase beta alone during Period 1; all patients were co-administered 150 mg migalastat HCl 2 hours prior to initiation of agalsidase beta during Period 2.
- Two patients (identified as Subjects A and B) received iv infusions of agalsidase beta 1.0 mg/kg for 2-hr durations.
- Five patients (identified as Subjects C, D, E, F, and G) received iv infusions of agalsidase beta 0.5 mg/kg for 2-hr durations with 1 exception: Subject E was infused 2 hrs and 40 min during Period 2, but was infused for 2 hours during Period 1.
- All subjects were males with Fabry Disease aged 44-61 year, body mass index (BMI) ranged from 20.9-29.1 kg/m^2 and estimated CLcr ranged from 54-88 mL/min. The genotype for each subject is presented in Table 1.

Table 1. *GLA* Genotypes

Subject ID	Genotype
Subject A	N298S
Subject B	717delAA
Subject C	N/A
Subject D	Q221X
Subject E	W236X
Subject F	R227X
Subject G	R227X

N/A = Not available

Safety

To date, 12 adverse events (AEs) have been reported, one of which was serious. The serious AE was a transient ischemic attack (TIA) which occurred after the screening visit, but prior to dosing, was moderate in severity, required hospitalization, and was considered unrelated to study drug by the investigator. The TIA resolved without sequalae. All other AEs were mild in severity, all considered unrelated to study drug, and most resolved without treatment. Three AEs in 3 different subjects are ongoing: premature atrial contractions, atrial flutter, and lower extremity edema, all unrelated to study drug.

Plasma α-Gal A Activity

Figure 1. Plasma AUC α-Gal A Activity vs. Treatment with 1.0 mg/kg Agalsidase Beta with Inserted Mean (SD) Activity-Time Profiles

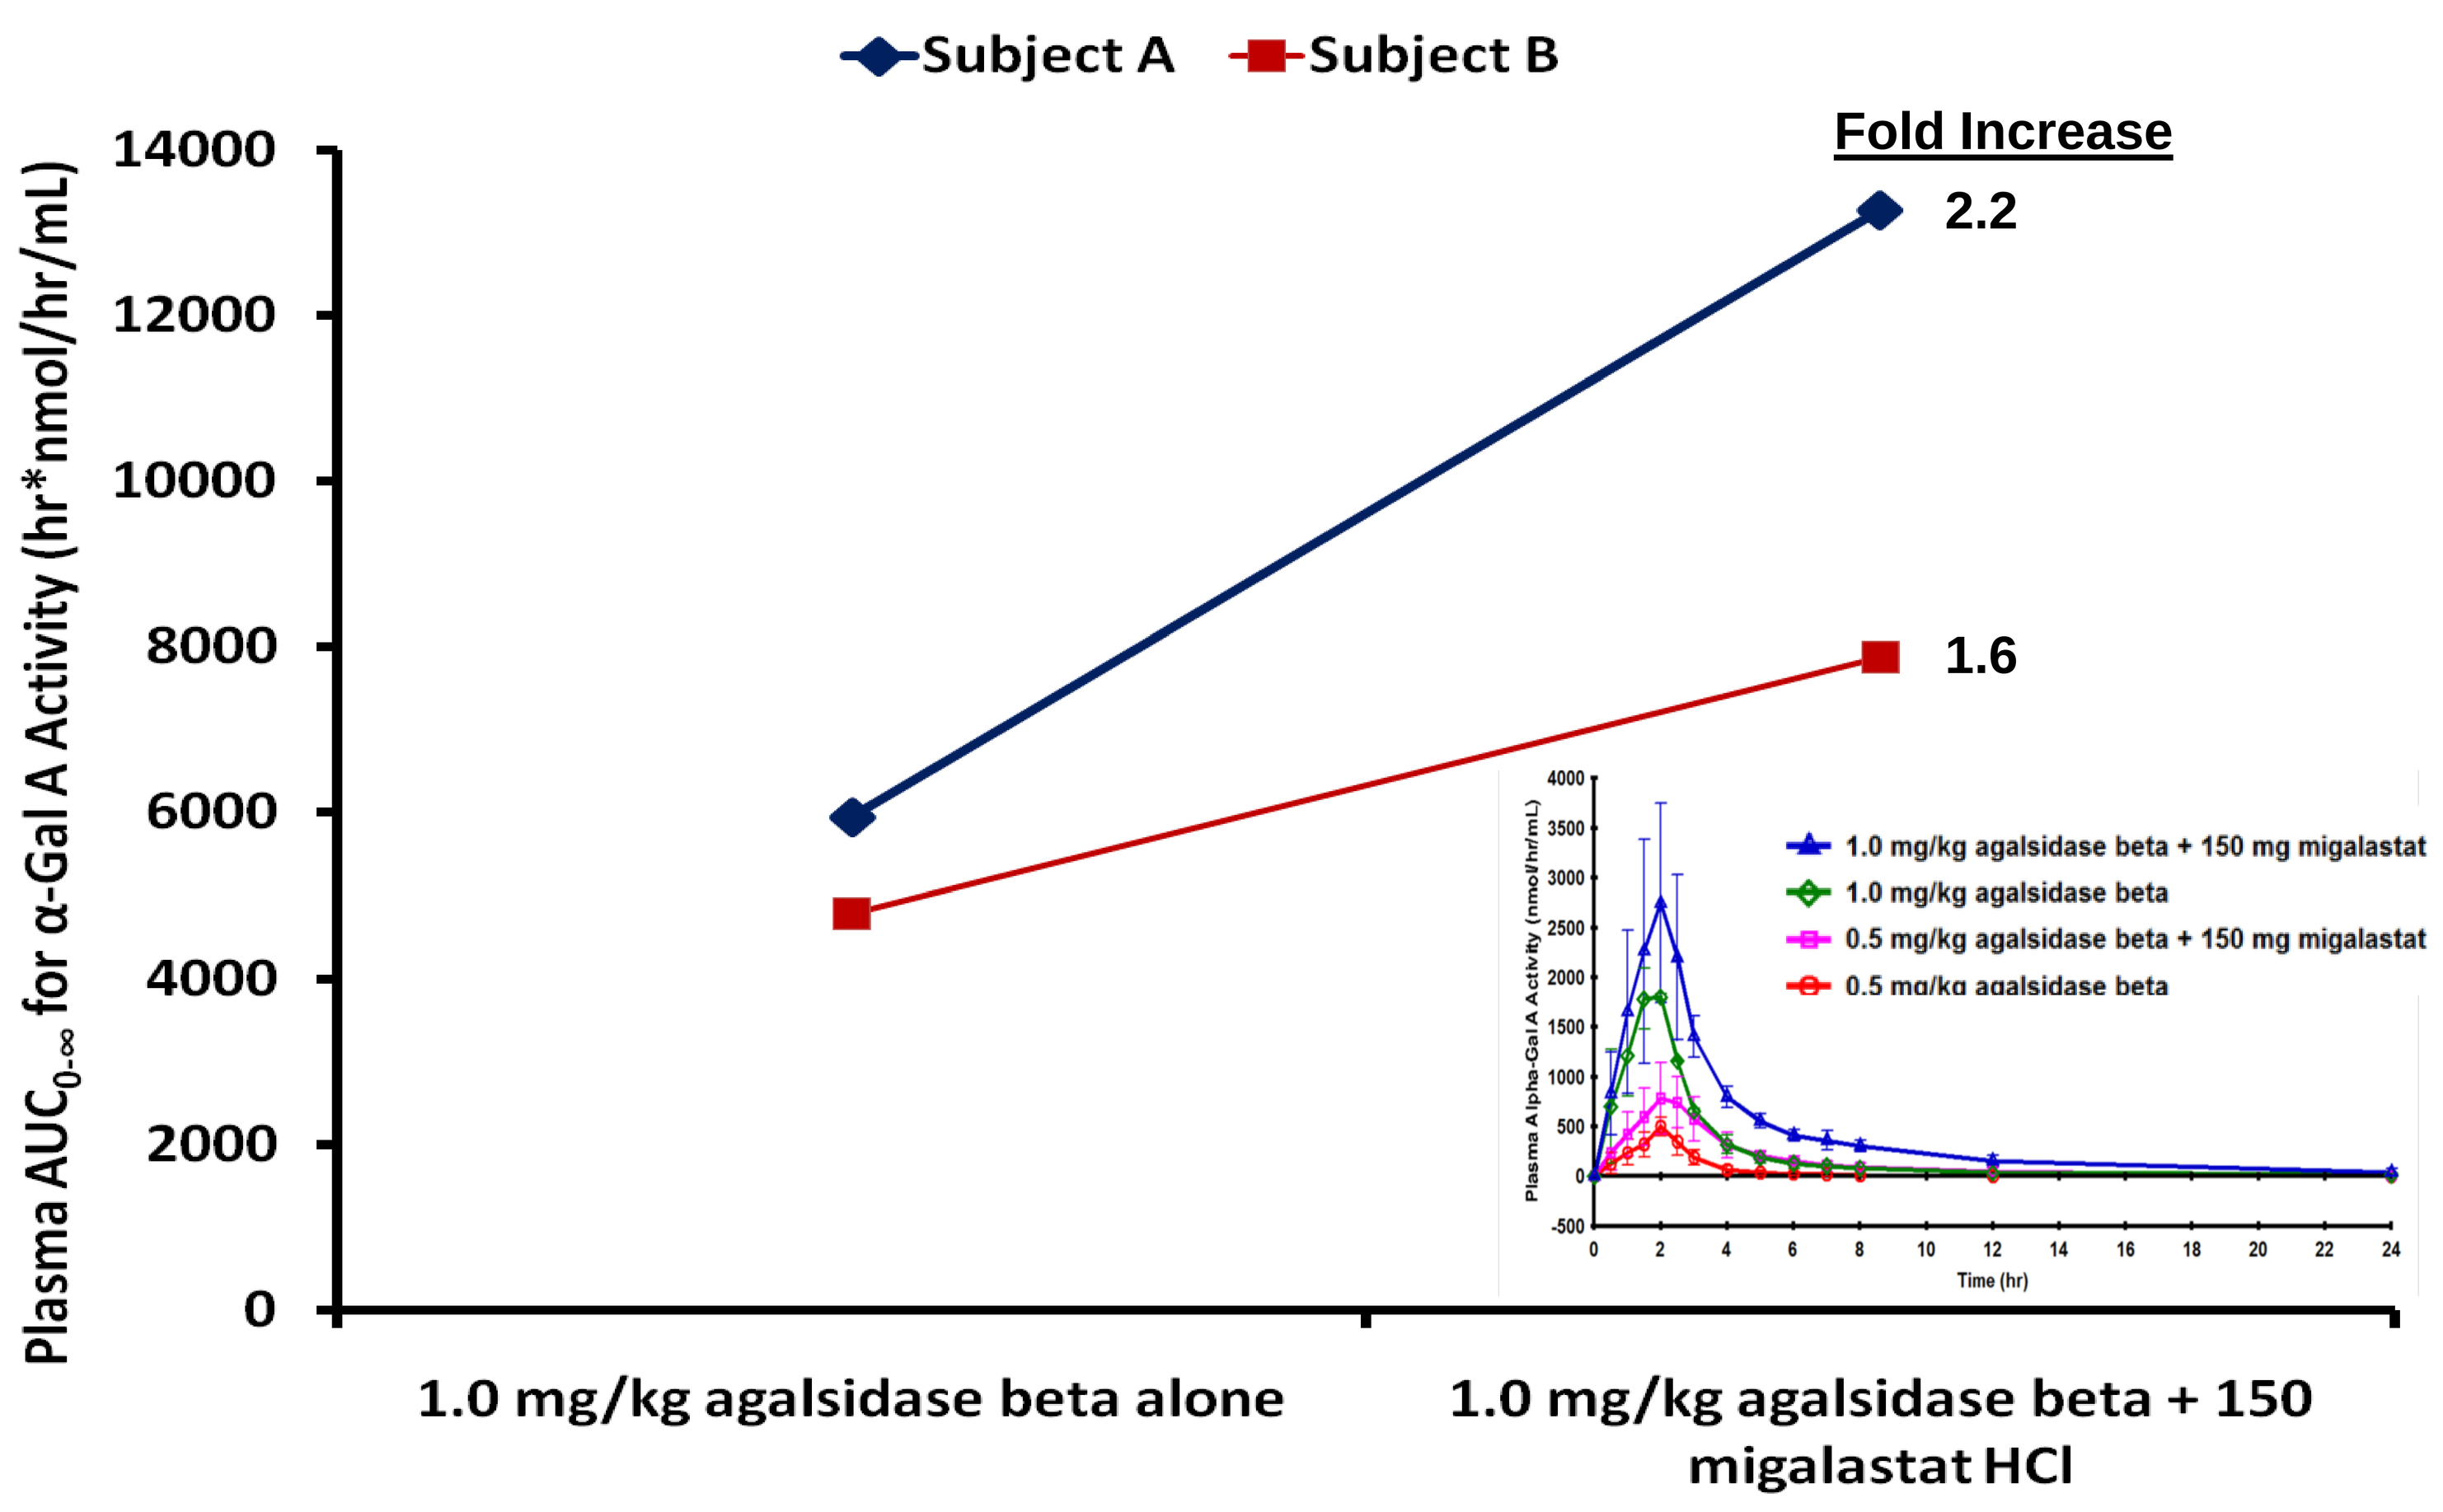
Subject A
Subject B
Fold Increase
2.2
1.6
Plasma AUC0-∞ for α-Gal A Activity (hr*nmol/hr/mL)
14000
12000
10000
8000
6000
4000
2000
0
1.0 mg/kg agalsidase beta alone
1.0 mg/kg agalsidase beta + 150 migalastat HCl
1.0 mg/kg agalsidase beta + 150 mg migalastat
1.0 mg/kg agalsidase beta
0.5 mg/kg agalsidase beta + 150 mg migalastat
0.5 mg/kg agalsidase beta
Time (hr)

Figure 2. Plasma AUC α-Gal A Activity vs. Treatment with 0.5 mg/kg Agalsidase Beta

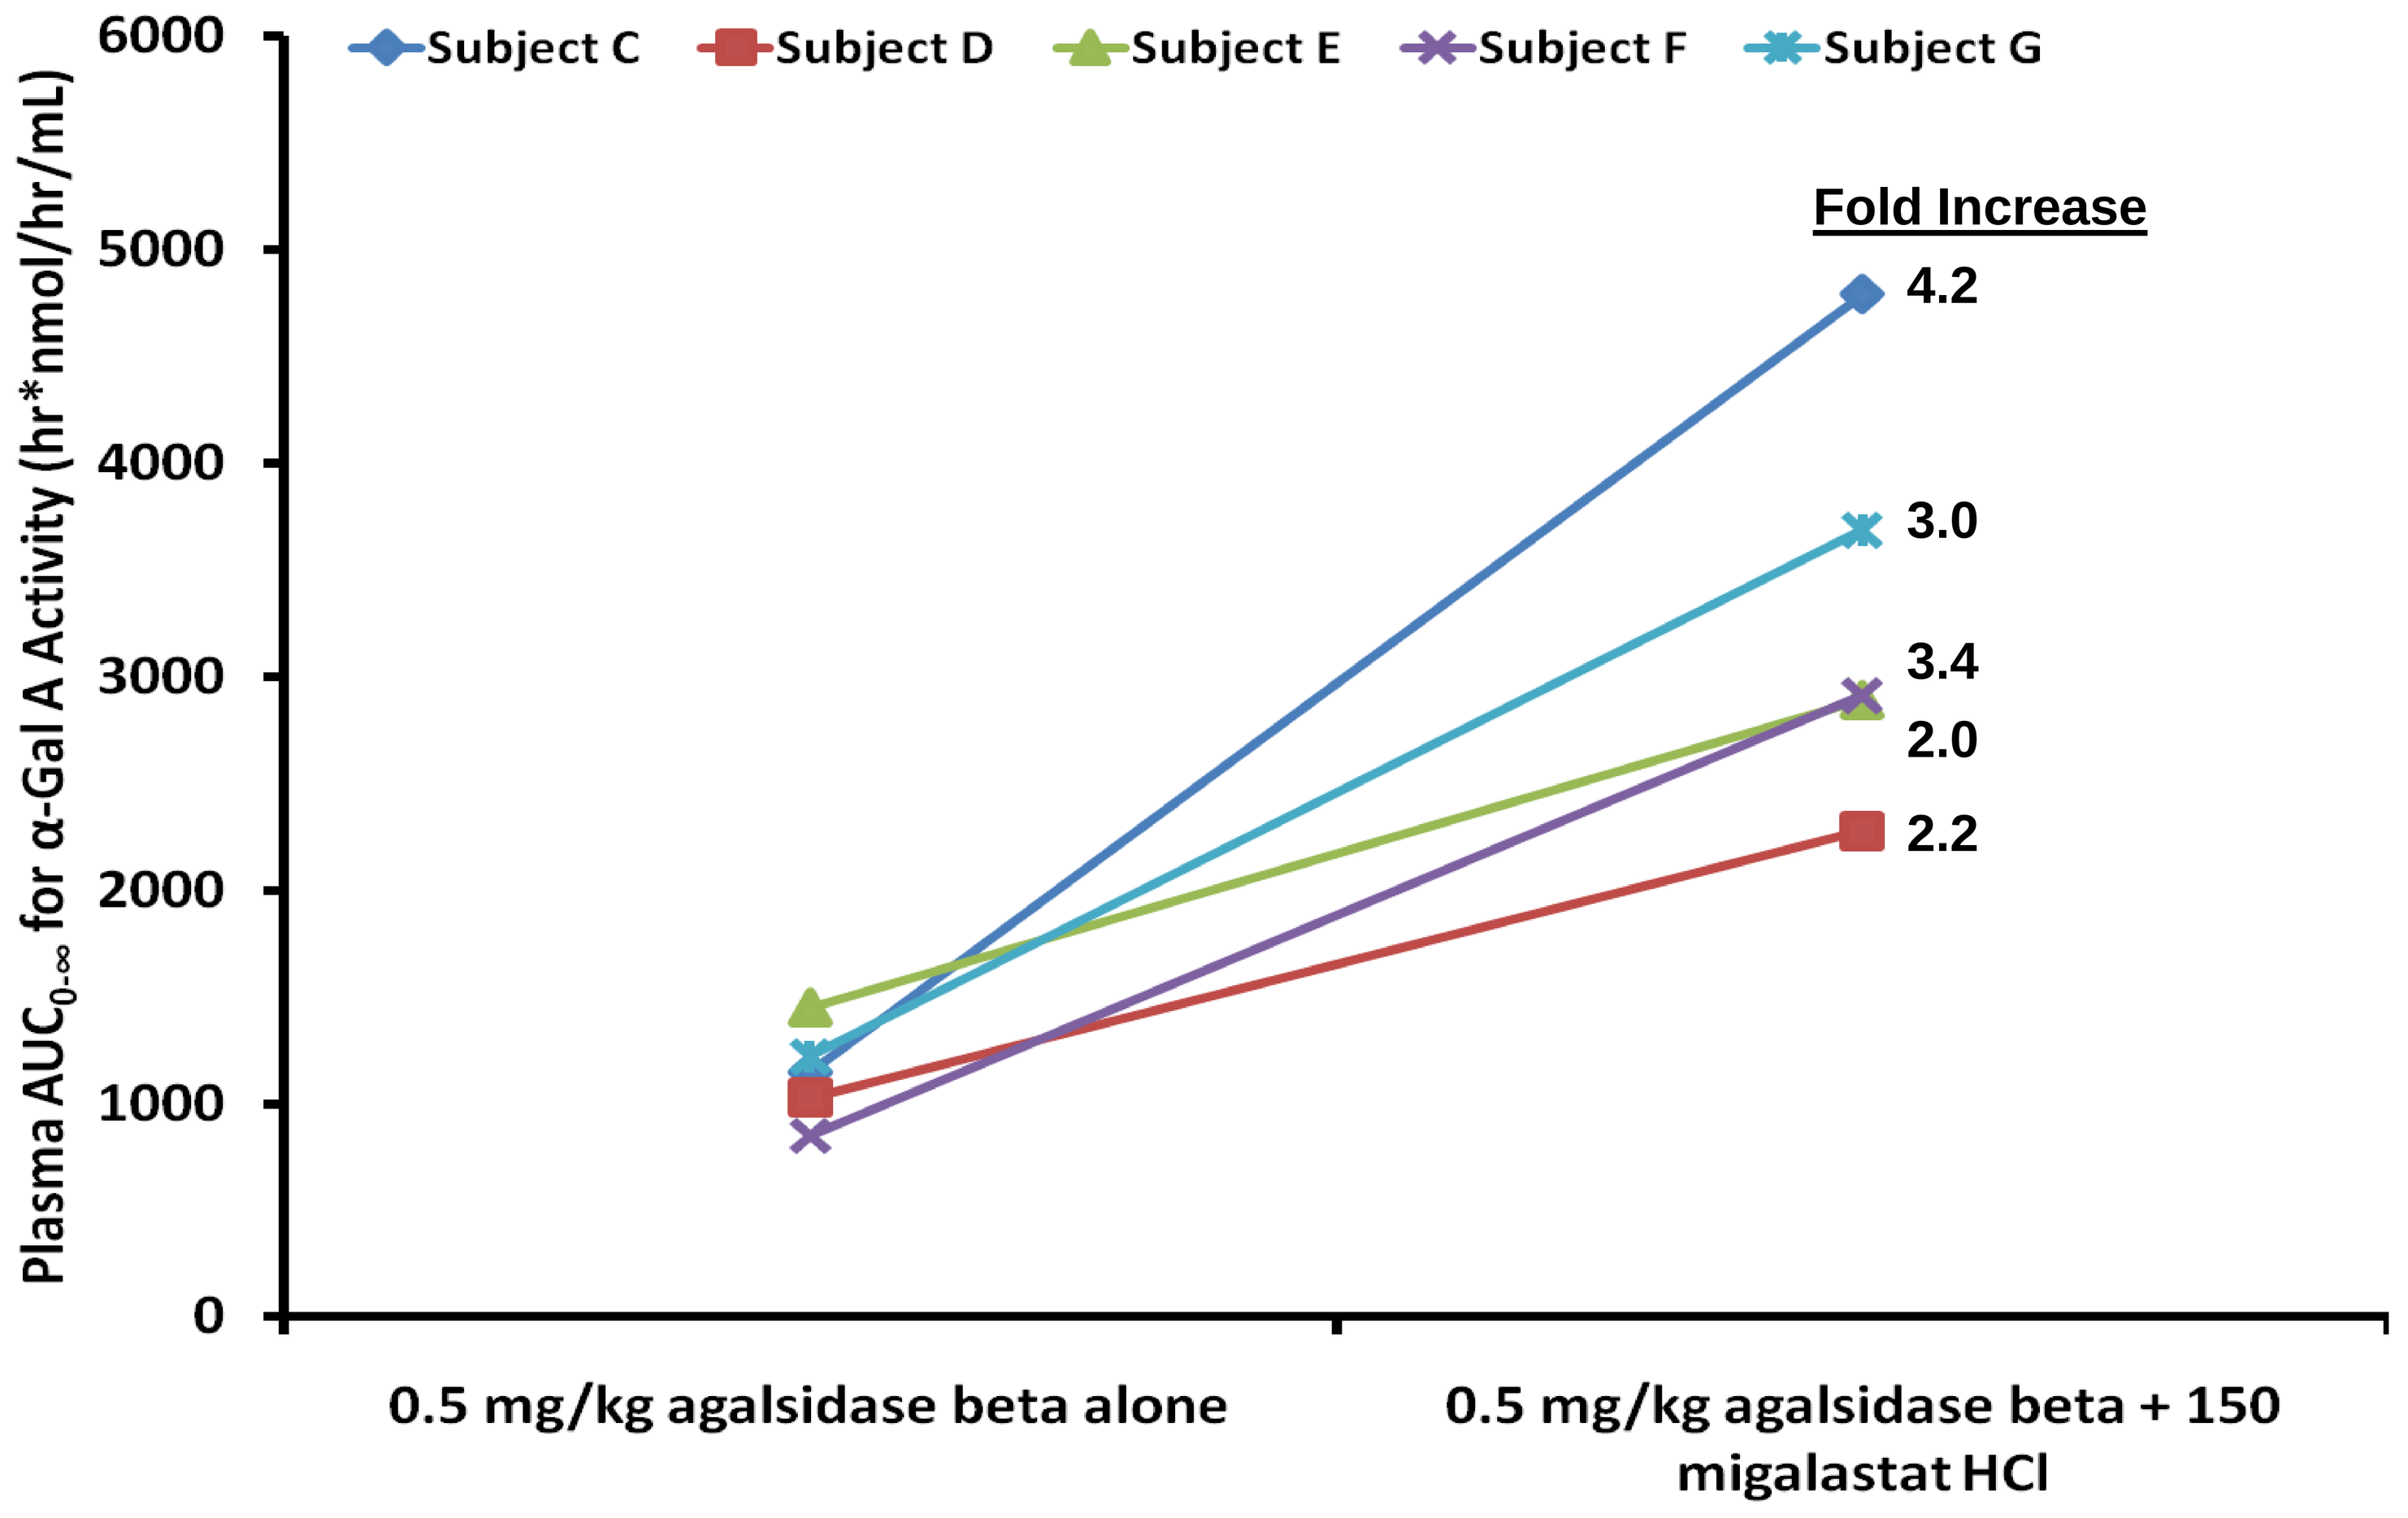
Subject C
Subject D
Subject E
Subject F
Subject G
Fold Increase
4.2
3.0
3.4
2.0
2.2
Plasma AUC0-∞ for α-Gal A Activity (hr*nmol/hr/mL)
6000
5000
4000
3000
2000
1000
0
0.5 mg/kg agalsidase beta alone
0.5 mg/kg agalsidase beta + 150 migalastat HCl

Skin α-Gal A Activity

Figure 3. Skin α-Gal A Activity on Day 2 with Baseline-subtracted Ratios from Agalsidase Beta Alone

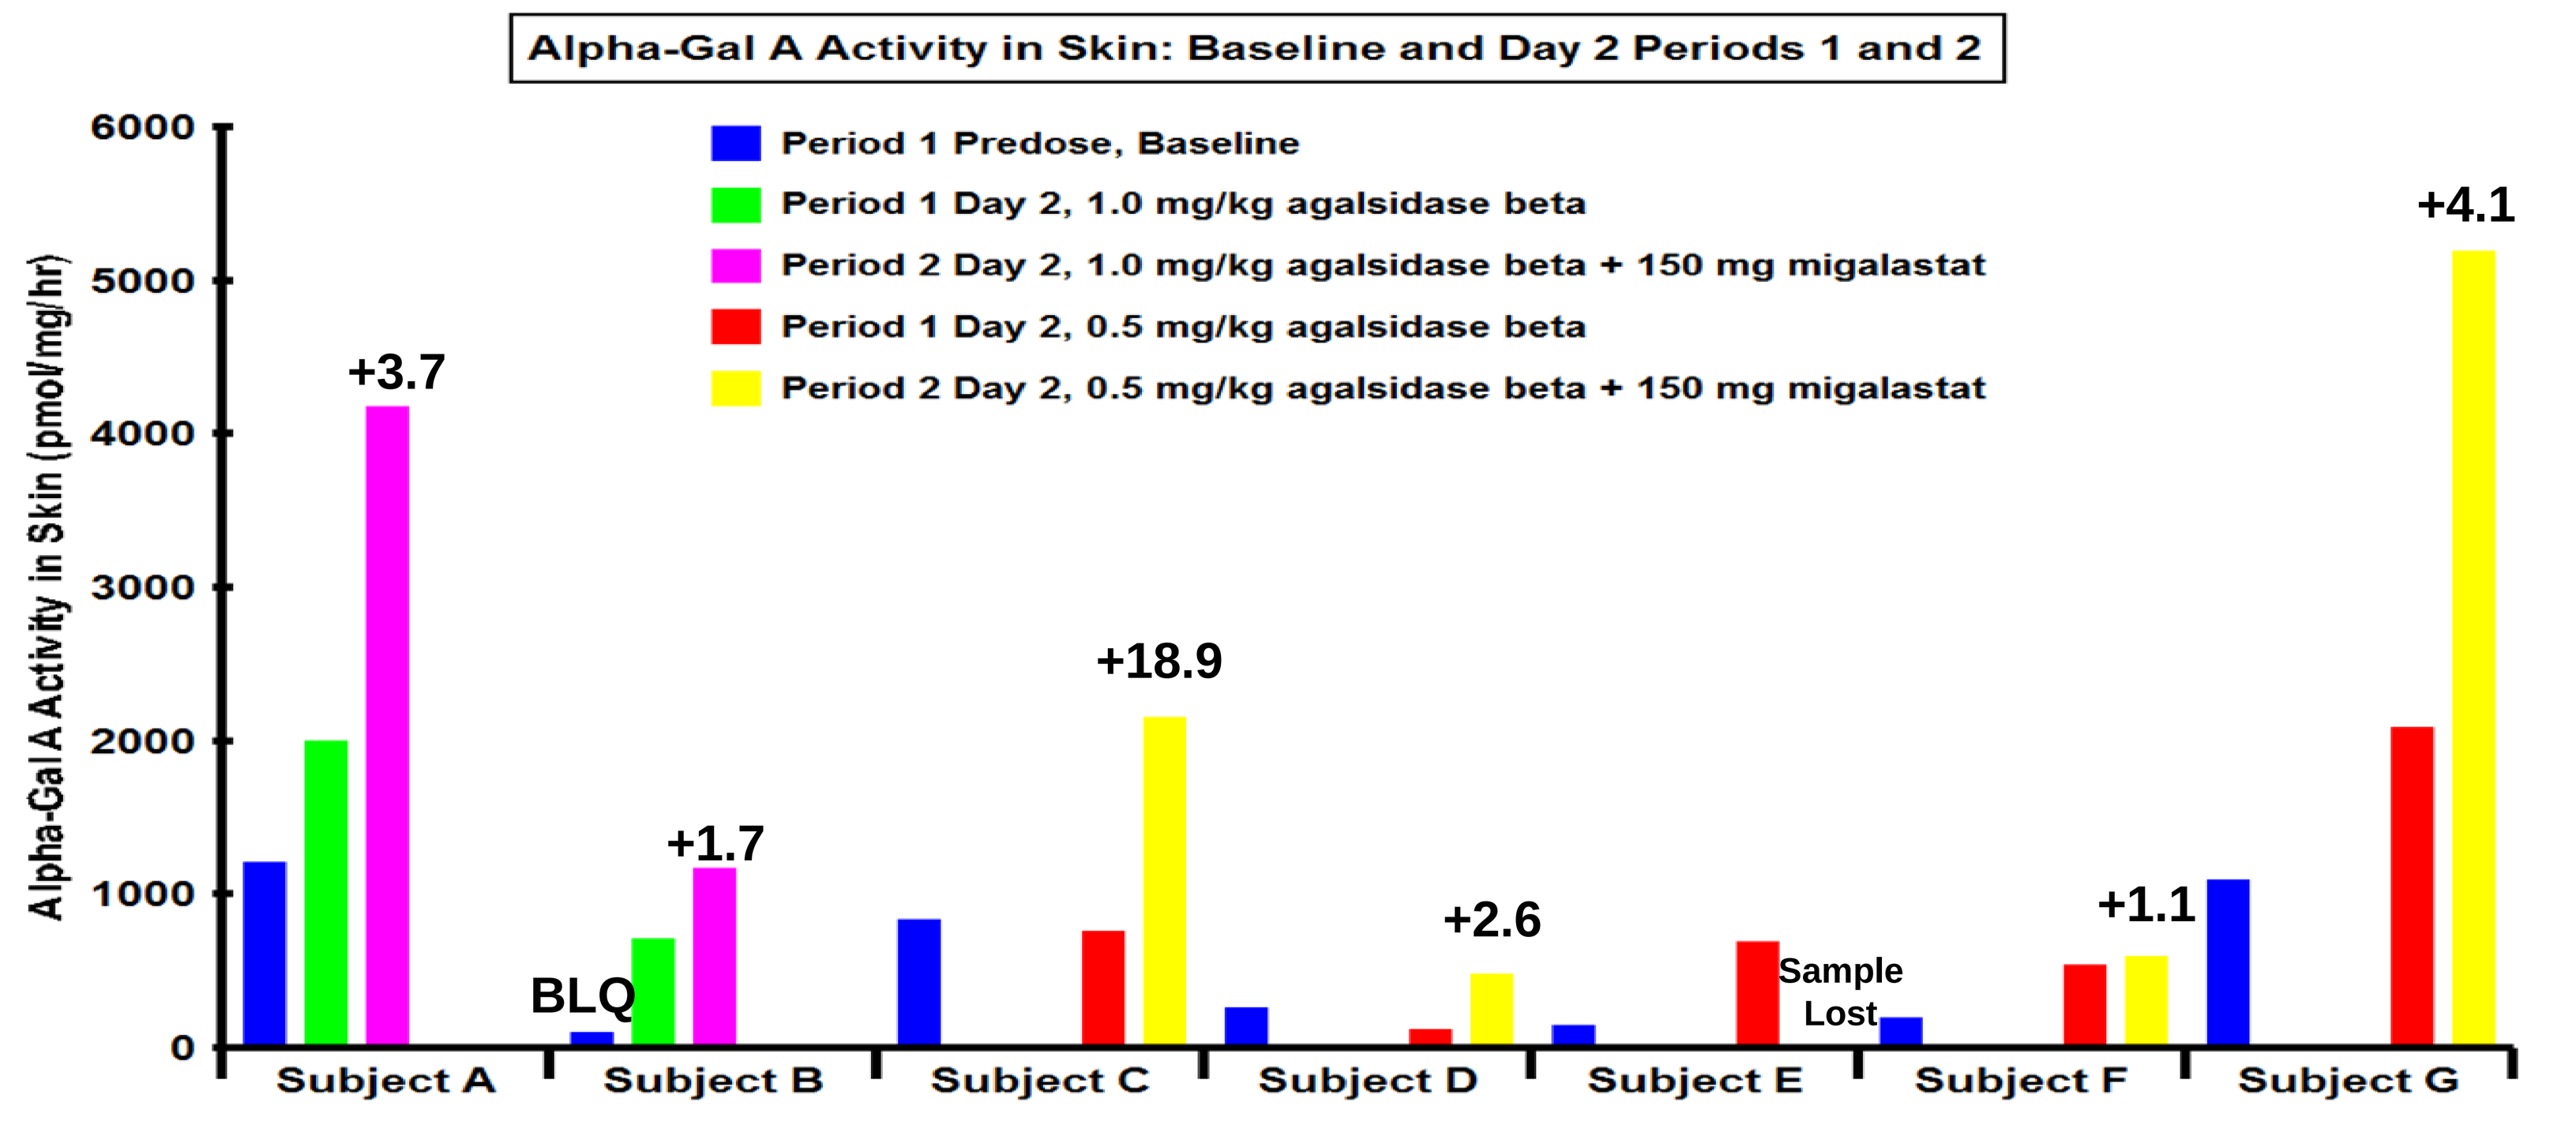
Alpha-Gal A Activity in Skin: Baseline and Day 2 Periods 1 and 2
Period 1 Predose, Baseline
Period 1 Day 2, 1.0 mg/kg agalsidase beta
Period 2 Day 2, 1.0 mg/kg agalsidase beta + 150 mg migalastat
Period 1 Day 2, 0.5 mg/kg agalsidase beta
Period 2 Day 2, 0.5 mg/kg agalsidase beta + 150 mg migalastat
Alpha-Gal A Activity in Skin (pmol/mg/hr)
6000
5000
4000
3000
2000
1000
0
+3.7
BLQ
+1.7
+18.9
+2.6
Sample Lost
+1.1
+4.1
Subject A
Subject B
Subject C
Subject D
Subject E
Subject F
Subject G

BLQ = Below the limit of quantitation; the lower limit of the analytical range was used as the baseline value for Subject B

Figure 4. Skin α-Gal A Activity on Day 7 with Baseline-subtracted Ratios from Agalsidase Beta Alone

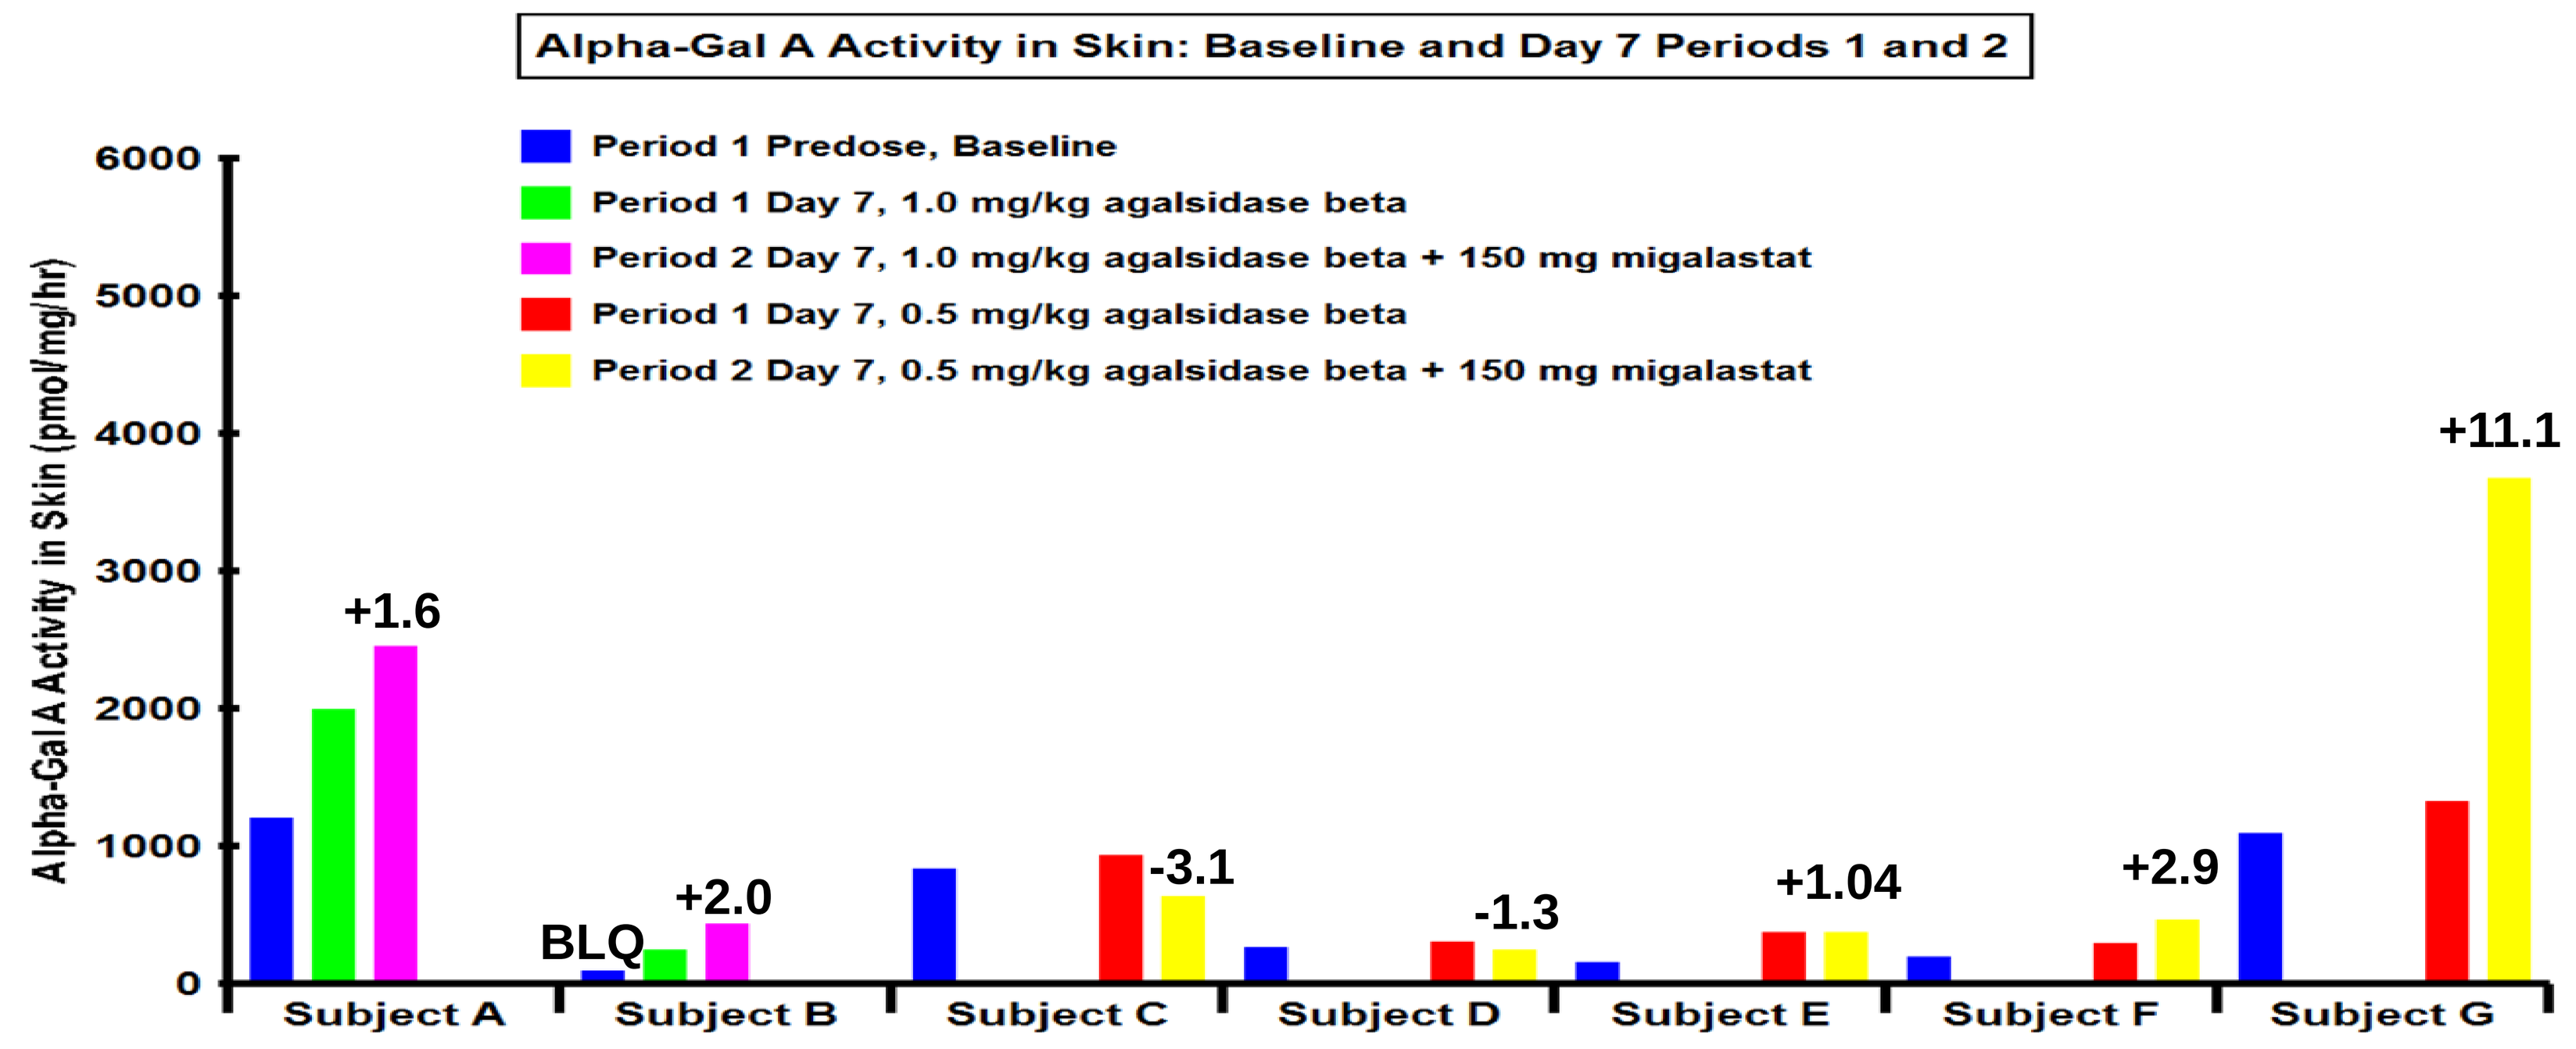
Alpha-Gal A Activity in Skin: Baseline and Day 7 Periods 1 and 2
Period 1 Predose, Baseline
Period 1 Day 7, 1.0 mg/kg agalsidase beta
Period 2 Day 7, 1.0 mg/kg agalsidase beta + 150 mg migalastat
Period 1 Day 7, 0.5 mg/kg agalsidase beta
Period 2 Day 7, 0.5 mg/kg agalsidase beta + 150 mg migalastat
Alpha-Gal A Activity in Skin (pmol/mg/hr)
6000
5000
4000
3000
2000
1000
0
+1.6
BLQ
+2.0
-3.1
-1.3
+1.04
+2.9
+11.1
Subject A
Subject B
Subject C
Subject D
Subject E
Subject F
Subject G

BLQ = Below the limit of quantitation; the lower limit of the analytical range was used as the baseline value for Subject B

Summary of Results

Plasma α-Gal A activity increased at all time points for most subjects for co-administration relative to enyzme replacement therapy (ERT) alone with one exception. Subject E received an unbalanced infusion, 40 minutes longer during period 2 which caused relative decreased enzyme activity during the infusion phase. However, all Subject E time points post peak activity were increased relative to ERT alone. Additionally, consistent with Eng et al[1], exposures increased in a non-linear manner for α-Gal A activity.

For co-administration with migalastat HCl (Period 2) relative to ERT alone (Period 1), all subjects had increased plasma α-Gal A activity AUC as observed in Figures 1 and 2. The mean increase in plasma α-Gal A activity AUC was 3.0-fold for 0.5 mg/kg agalsidase beta, excluding 2.0-fold patient with the unbalanced infusion duration, the mean increase was 3.2-fold. The mean increase in plasma α-Gal A activity AUC was 1.9-fold for 1.0 mg/kg agalsidase beta.

The following mean increases in skin α-Gal A activity were observed: 2.6-fold and 1.4-fold on Days 2 and 7, respectively, for 0.5 mg/kg agalsidase beta (N=5), and 1.9- and 1.5-fold on Days 2 and 7, respectively, for 1.0 mg/kg agalsidase beta (N=2).

Although the relevance of α-Gal A activity in PBMCs is currently unknown, the following mean increases in PBMC α-Gal A activity were observed : 2.4-, 1.9-, and 2.1-fold on Days 2, 7, and 14, respectively, for 0.5 mg/kg agalsidase beta (N=5), and 1.8-, 4.8- and 3.5-fold for 1.0 mg/kg (N=2) agalsidase beta on Days 2, 7 and 14, respectively.

No change observed in plasma α-Gal A protein by Western Blot Period 2 (co-administration) / Period 1 (ERT alone) AUC ratio for 5 out of 7 subjects to date, however, 2 subjects, (Subject C who received 0.5 mg/kg agalsidase beta and Subject G who receive 1.0 mg/kg agalsidase beta) had 20% increases following co-administration relative to ERT alone.

Preliminary Conclusions

Preliminary results suggest 150 mg migalastat interaction with 0.5 mg/kg and 1.0 mg/kg agalsidase beta resulted in α-Gal A activity increases in plasma, skin, and PBMCs confirming observations from preclinical studies.

Co-administration of 150 mg migalastat HCl with agalsidase beta was generally safe and well-tolerated.

Further investigations will be conducted in the current study with the 150 mg migalastat HCl dose and agalsidase alpha and the 450 mg dose of migalastat HCl (Stage 2) with agalsidase alpha and beta.

References: 1. Eng, C, et al. A phase 1/2 clinical trial of enzyme replacement in Fabry Disease: pharmacokinetic, substrate clearance, and safety studies. *Am. J. Hum. Genet. 68:711-722, 2001*.