LP-048

A Survey of Patient Registries in Epidermolysis Bullosa: A Call for Standardized Methodology and Increased Collaboration

Bodemer C[1], Paller A[2], Wisk J[3], Giuliano J[3], Cantor E[3], Lagast H[3]

[1]The University Hospital Necker Enfants Malades, Paris, France; [2]Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [3]Amicus Therapeutics, Inc., Cranbury, NJ, USA

INTRODUCTION

- Epidermolysis bullosa is a rare, often severe, genetic disorder characterized by mechanical fragility and blistering or erosion of the skin, mucosa, or epithelial lining of other organs in response to little or no apparent trauma
- Epidermolysis bullosa can lead to severe multisystem complications and, sometimes, premature death
- There are no approved treatments for epidermolysis bullosa
- Many uncertainties remain in the understanding of the disease, including natural disease history and genotype-phenotype relationships
 - To address this knowledge gap, multiple epidermolysis bullosa disease registries have been developed in recent decades, which differ widely in strategy and scope

Registries

- In rare diseases, the small number of patients makes it challenging to understand many aspects of the disease
- Patient registries, which systematically collect a uniform set of data in patients with a particular condition or exposure, can facilitate medical and scientific research
- Data input may be physician or patient reported and can include a variety of information and outcomes
- Two main types of registries exist in the medical community
 - In a disease or condition registry, patients are included if they have a specific disease
 - In a product or treatment registry, data are collected from patients who have been exposed to a particular product (ie, drug or device), and the registry may be required by regulatory agencies after approval of a new therapeutic agent

OBJECTIVE

- To better understand the current state of epidermolysis bullosa registry work and to determine the quality of existing registries using accepted registry standards, a comprehensive survey of the epidermolysis bullosa registry landscape was undertaken

METHODS

Registry Search

- Registries were identified using an online search conducted between December 2016 and February 2017
 - The search strings "Epidermolysis Bullosa AND registry" and "Epidermolysis Bullosa AND register" were used to search PubMed and Google
 - The first 100 results from the Google search and all results from the PubMed search were analyzed
 - The term "Epidermolysis Bullosa" was searched on ClinicalTrials.gov; all results were analyzed
 - The reference lists of all pertinent search results were also investigated

Registry Selection

Registry Inclusion Criteria

- The following criteria were used to include epidermolysis bullosa patient registries in this review:
 - The study collected and stored uniform data on patients with epidermolysis bullosa
 - Data collection was systematic and at least partly active (ie, data were collected for the purpose of the registry)
 - Study design was observational
 - The registry was described by an accessible publication or website

Registry Selection

- The search identified 30 studies that might represent epidermolysis bullosa registries (**Figure 1**)
 - Nine registries with inaccessible publications or websites were excluded, resulting in 21 studies that provided sufficient information for further characterization
 - Of these 21 studies, 4 were excluded because of the lack of active data collection (ie, review of records or hospital database), resulting in 17 registries for analysis

Figure 1. Registry Search and Selection

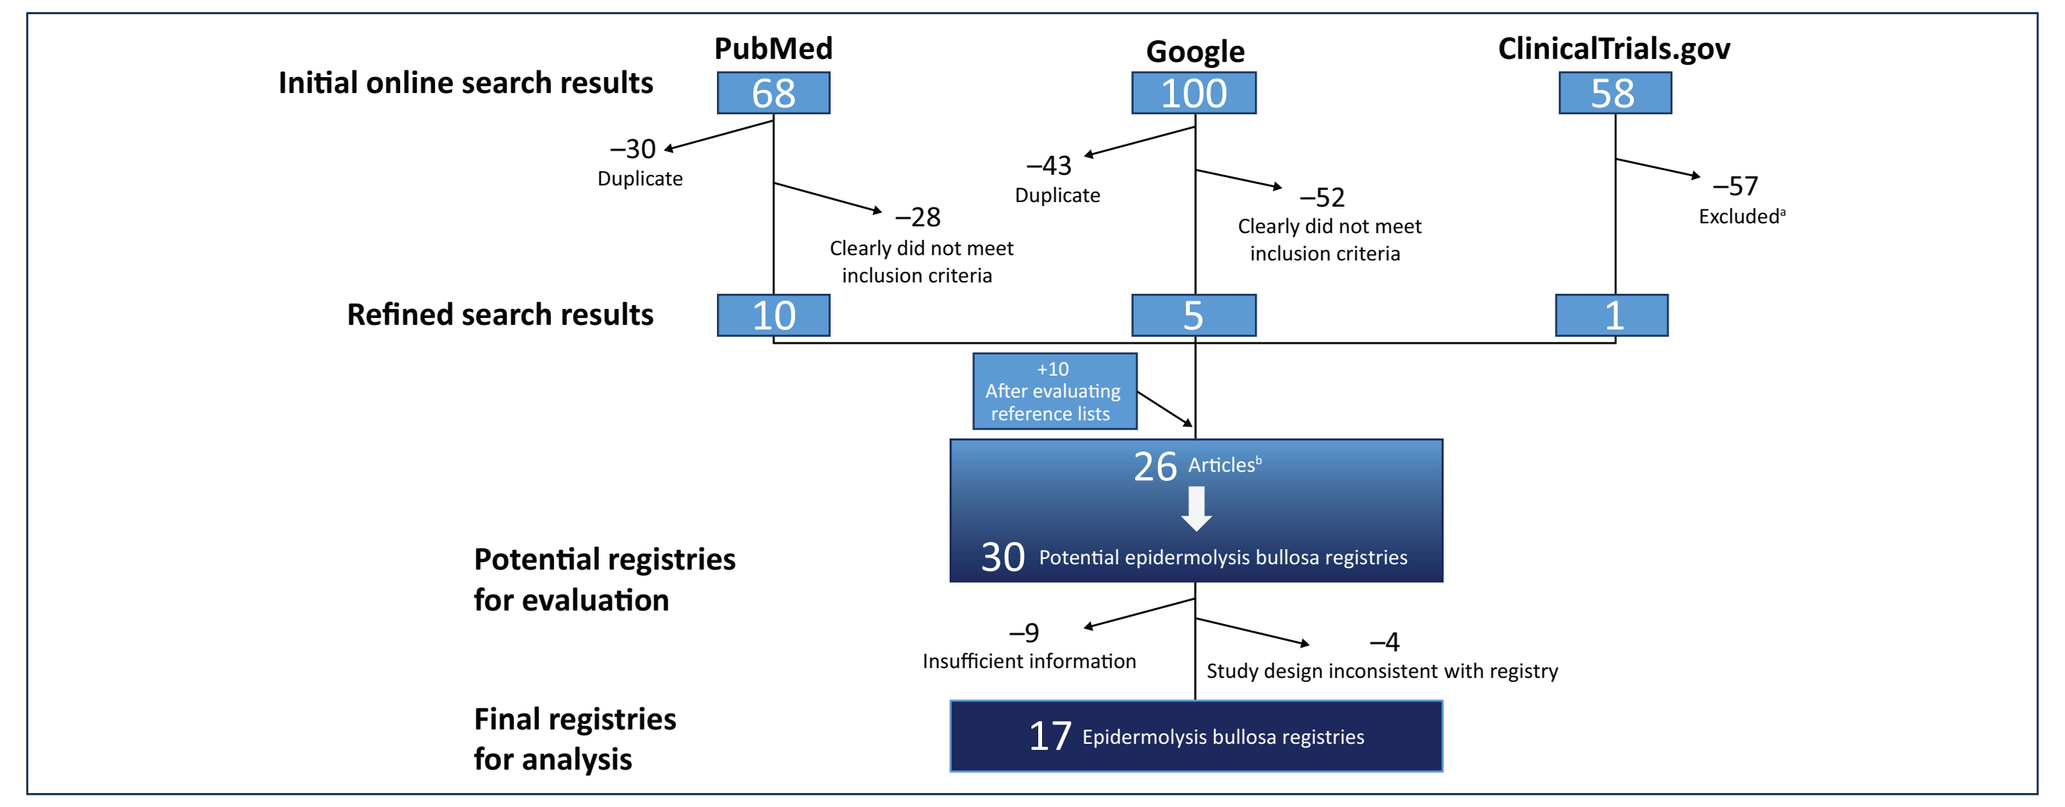
Initial online search results
PubMed
68
Google
100
ClinicalTrials.gov
58
−30
Duplicate
−28
Clearly did not meet inclusion criteria
−43
Duplicate
−52
Clearly did not meet inclusion criteria
−57
Excluded[a]
Refined search results
10
5
1
+10
After evaluating reference lists
26 Articles[b]
30 Potential epidermolysis bullosa registries
Potential registries for evaluation
−9
Insufficient information
−4
Study design inconsistent with registry
Final registries for analysis
17 Epidermolysis bullosa registries

[a]Reasons for exclusion: Interventional study (n=43), study did not involve hereditary epidermolysis bullosa (ie, epidermolysis bullosa acquisita) (n=4), study withdrawn (n=4), other observational study (n=4), duplicate (n=2).
[b]Some articles described multiple registries.

THE IDEAL REGISTRY

- The ideal patient registry includes 5 critical components that, when working together, will maximize the ability of the registry to contribute knowledge to the patient and medical communities (**Figure 2**)

Figure 2. Five Components of an Ideal Registry[a]

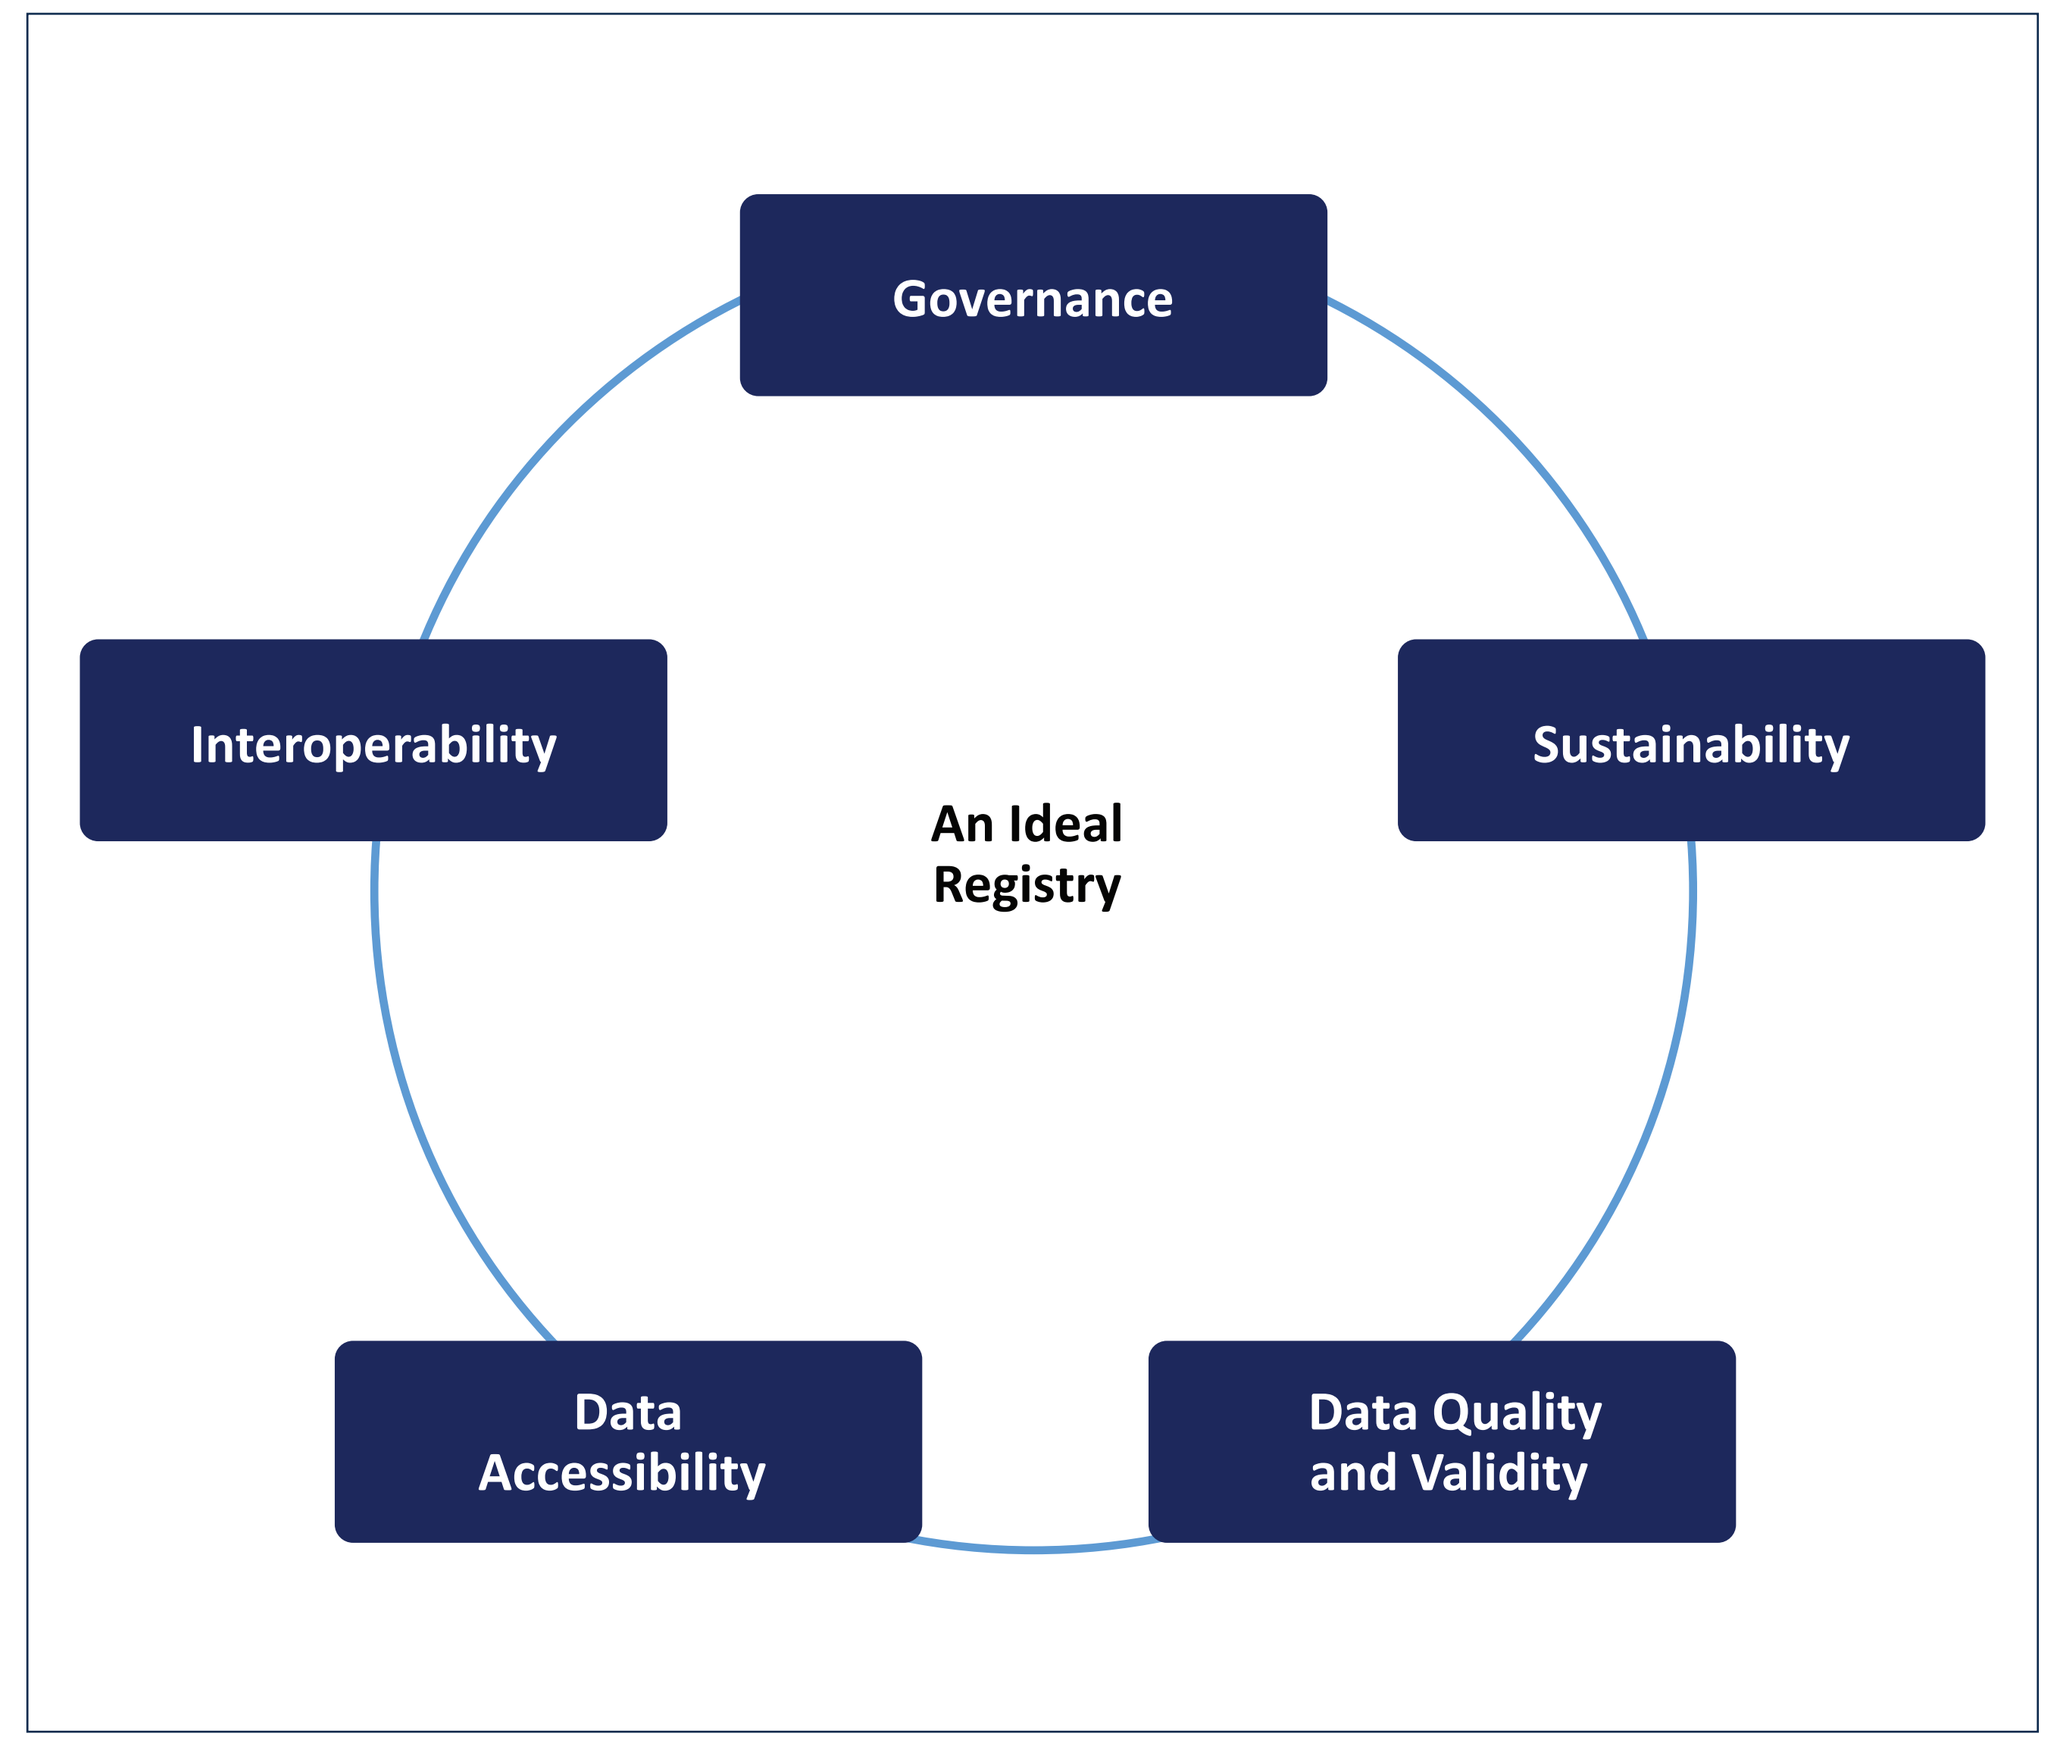
Governance
Interoperability
Sustainability
An Ideal Registry
Data Accessibility
Data Quality and Validity

[a]Coi A et al. *Public Health Genomics*. 2016;19(2):108-115.

Governance

- *Governance:* Governance represents the organized decision making and oversight of a registry
 - A steering committee (single or multiple committees) should include multiple stakeholders from various backgrounds, including patient representatives. This committee functions throughout the life cycle of a registry, including design of the study, oversight of registry operation, and reporting of data
- Overall, registry governance was poorly described among the publications and websites examined. It is unclear whether this represents a lack of governance structure in the creation and operation of epidermolysis bullosa registries or underreporting of such a structure
- An organized steering committee was identified in at least 1 registry; however, this information was not included in the associated publication and was found in a separate source

Sustainability

- *Sustainability:* Long-term funding and staffing are critical for the continued operation of the registry
- Across all registries, there was a wide range of geographic scope, registry size, and study duration (**Figure 3**)
 - The smallest registry included 58 patients and the largest included 3280 patients
 - Duration of the registries ranged from 1 year to 28 years (median, 7 years), and 7 registries (41%) are ongoing
- Funding sources of the registries were mixed and often of multiple resources
 - Most registries were funded by government or patient advocacy organizations (7 registries each); 5 registries were privately funded and 4 were funded by universities

Figure 3. (A) Geographic Scope, (B) Enrollment Size, and (C) Duration of Available Epidermolysis Bullosa Patient Registries (N=17)

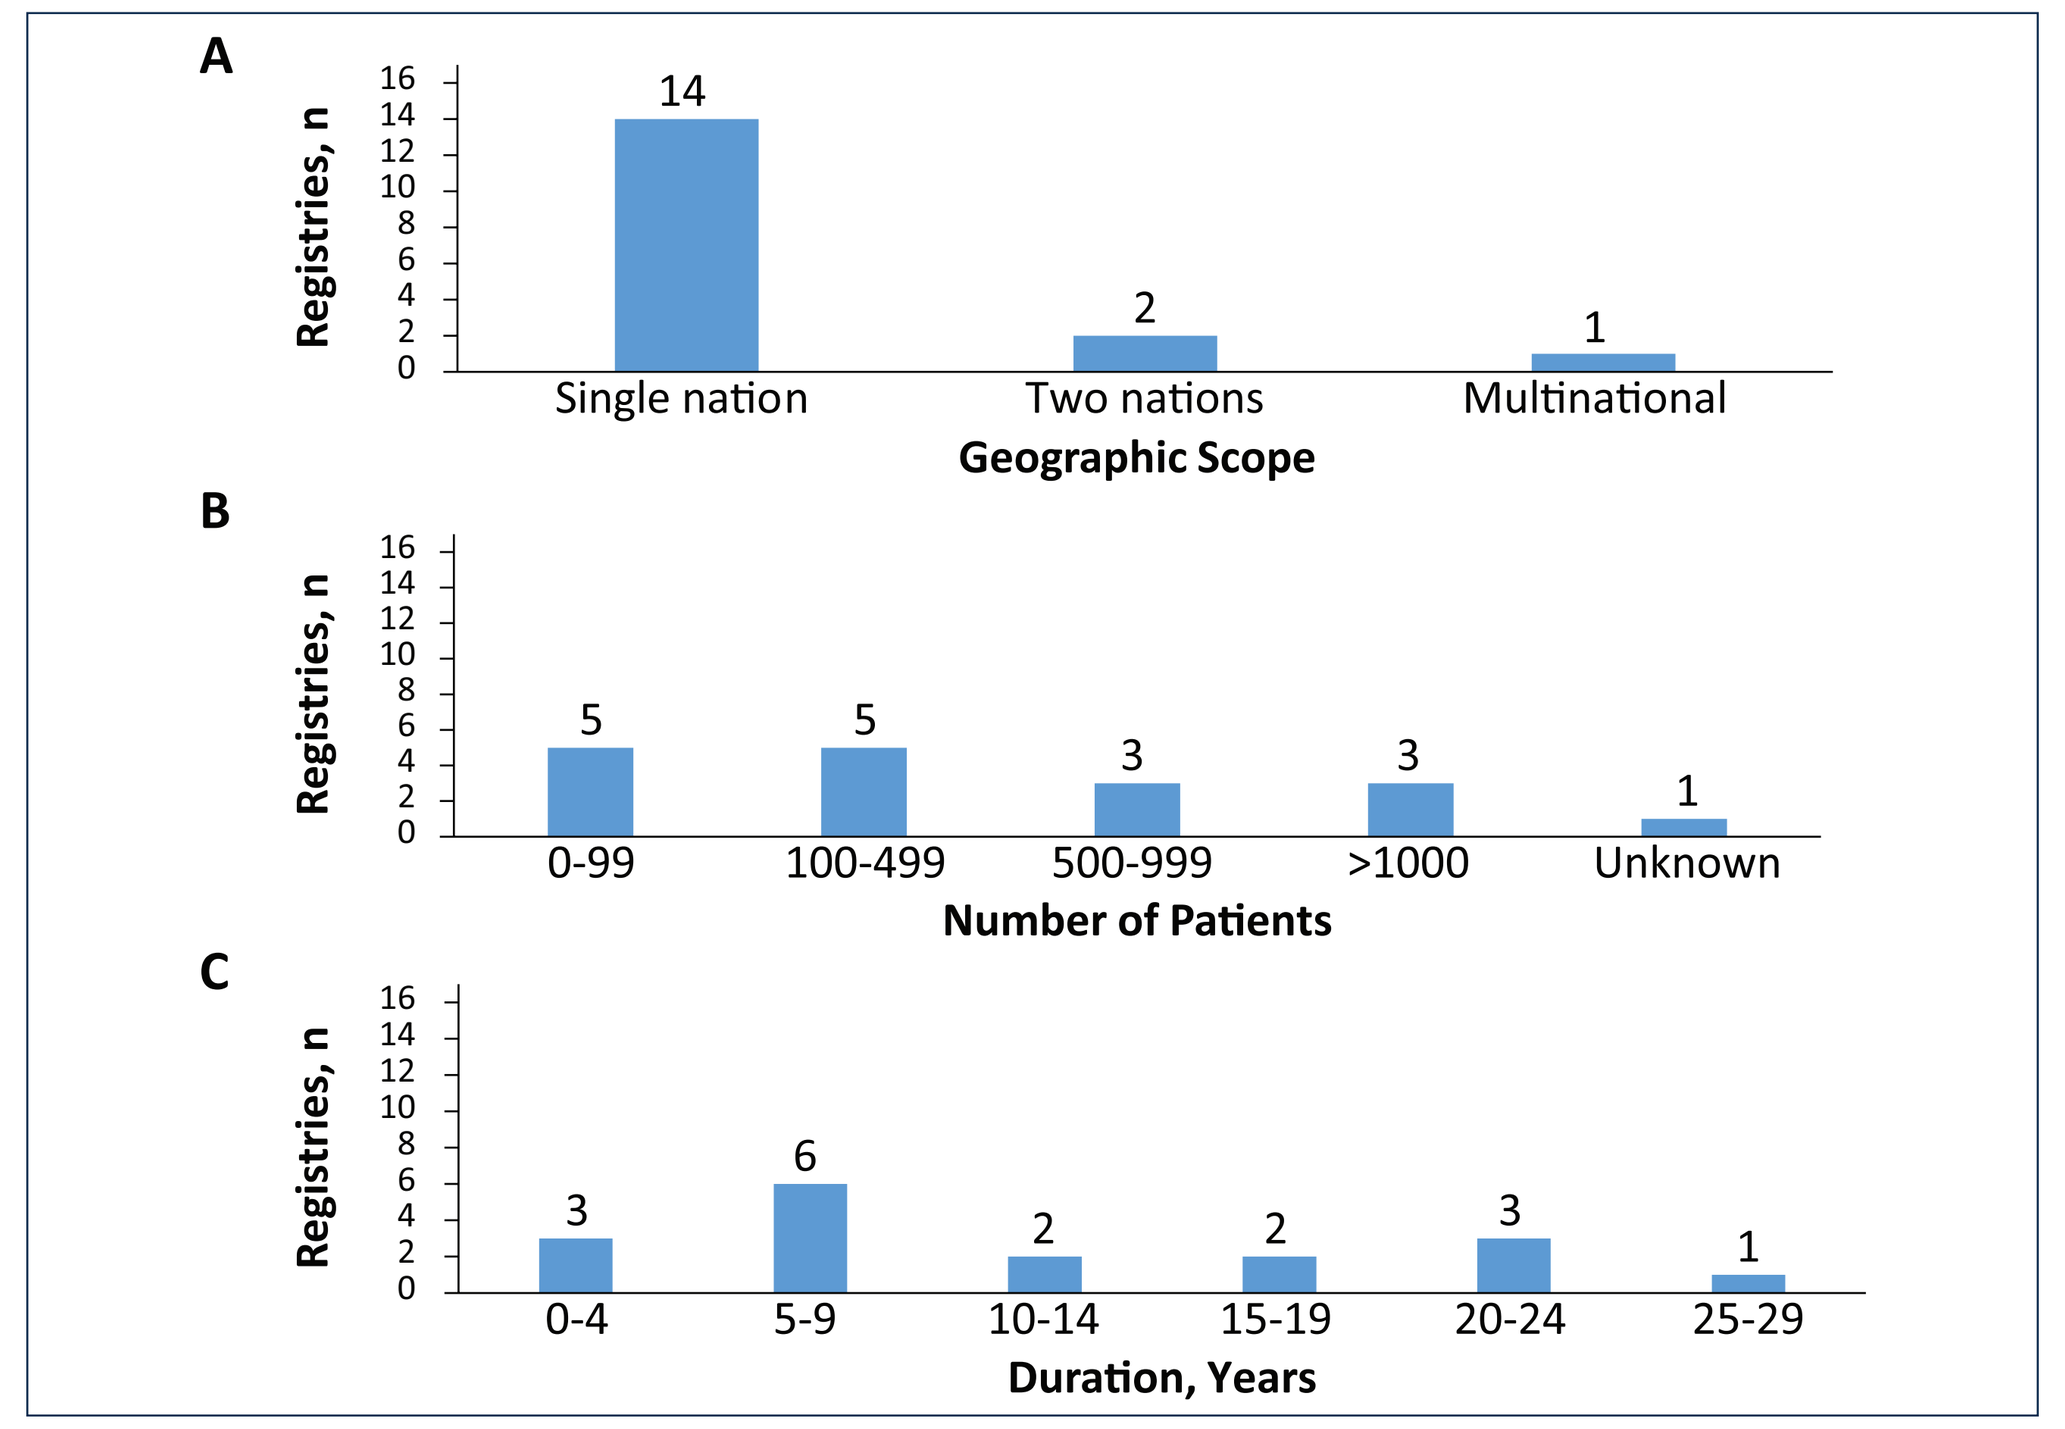
A
Registries, n
14
2
1
Single nation
Two nations
Multinational
Geographic Scope
B
Registries, n
5
5
3
3
1
0-99
100-499
500-999
>1000
Unknown
Number of Patients
C
Registries, n
3
6
2
2
3
1
0-4
5-9
10-14
15-19
20-24
25-29
Duration, Years

Seven studies are ongoing, and as a result duration data are subject to change.

Data Quality and Validity

- *Data quality and validity:* Data must be of good quality, and content must be relevant to answer scientific questions. A smaller common data set should be implemented to promote 100% completion and support interoperability; this data set should be shared with other investigators
- Overall, data collected in epidermolysis bullosa registries are not uniform. For example, of the 17 registries, 3 (18%) required documentation of genotype information and 8 (47%) required documentation of direct clinical examination findings by a registry investigator (**Figure 4**). Only 1 registry clearly stated that direct clinical examination findings and genotype data in every patient enrolled were collected
- Only 2 (12%) registries collected patient-reported data as part of the registry questionnaire
- Data ascertainment methods also varied. Six registries (35%) described active recruitment of patients (eg, advertisements, communications to physicians), whereas most registries involved purely passive recruitment (eg, registering only existing clinic patients, patients within a hospital network, or evaluation of referrals to a center)
- Overall, registries did not specify methods of quality control of the data

Figure 4. Required Documentation of Selected Data Points of Available Epidermolysis Bullosa Patient Registries (N=17)

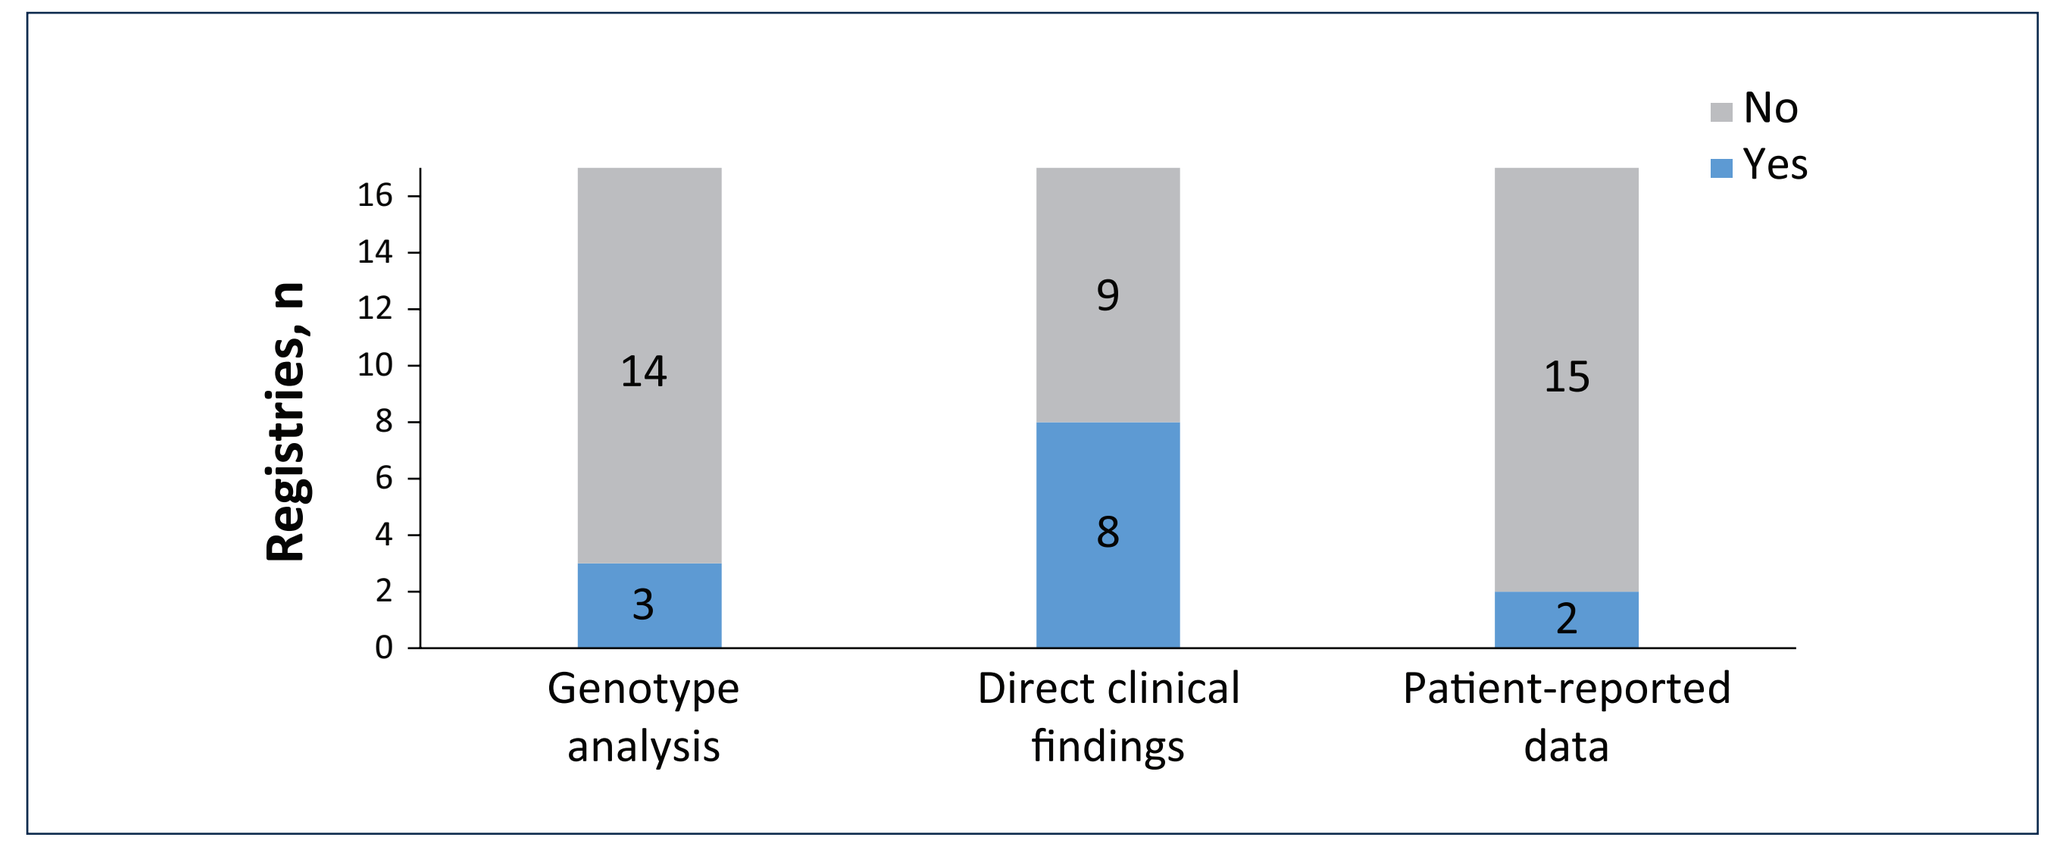
No
Yes
Registries, n
14
3
9
8
15
2
Genotype analysis
Direct clinical findings
Patient-reported data

Interoperability

- *Interoperability:* International collaboration is necessary to ensure that data are entered into common and compatible platforms, with consideration of data sharing and protection regulations in each country. A core data set with clearly defined definitions and ontology can be used to allow aggregation of registries that collect the same data points
- A common, ideally electronic, platform is critical for registry interoperability, so that data can be collected at multiple centers across multiple countries. In most registries, information regarding data entry was not sufficiently specific to further characterize; however, none of the registries referenced a common platform to another registry
- Another example of challenges in the interoperability of existing epidermolysis bullosa registries is the result of improved disease diagnosis leading to changes in epidermolysis bullosa classification. Major revisions in the classification scheme occurred as recently as 2008 and 2013. Among ongoing registries, it is not clear from the search results whether epidermolysis bullosa diagnosis nomenclature will be updated for patients enrolled before these dates
- Since initial classification of epidermolysis bullosa as a distinct entity in 1962, there have been 4 consensus classification meetings (1988, 1999, 2007, 2013) to reflect new disease understanding and technology (**Figure 5**)
- Initiation of the analyzed registries has occurred at different times, each with a different classification scheme for epidermolysis bullosa (**Figure 5**)

Figure 5. Timeline of Initiation Dates of Available Epidermolysis Bullosa Patient Registries in Relation to Epidermolysis Bullosa Reclassification Events (N = 17)

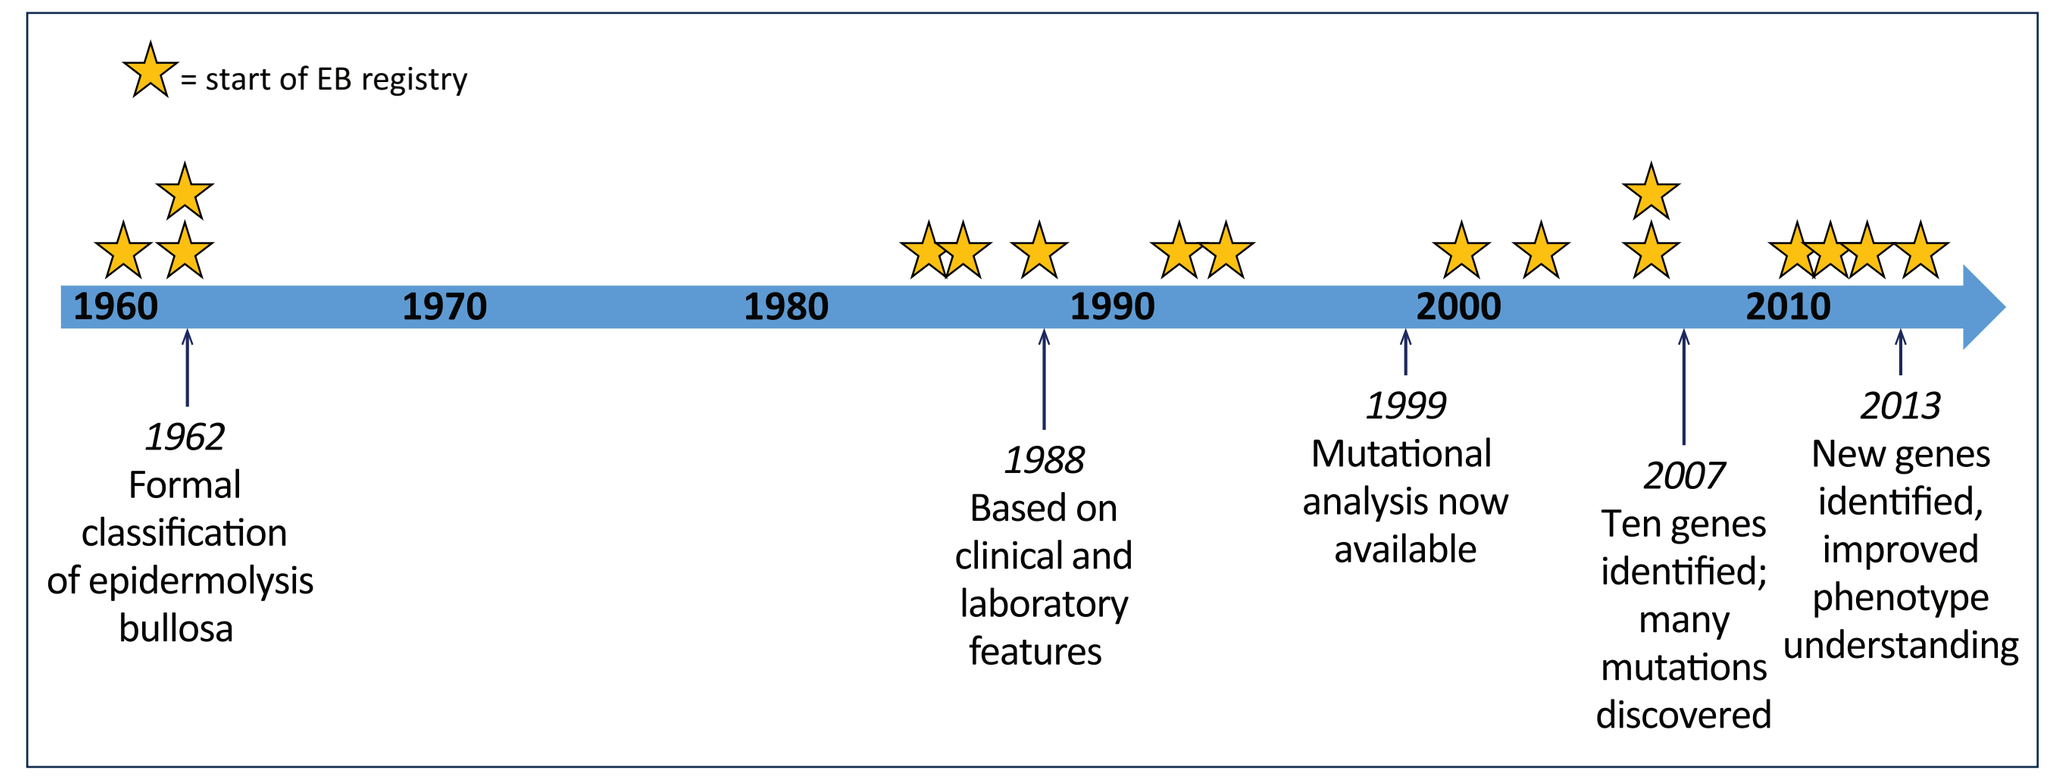
= start of EB registry
1960
1970
1980
1990
2000
2010
1962
Formal classification of epidermolysis bullosa
1988
Based on clinical and laboratory features
1999
Mutational analysis now available
2007
Ten genes identified; many mutations discovered
2013
New genes identified, improved phenotype understanding

The start date of 1 registry is unknown.

Data Accessibility

- *Data accessibility:* Once data are entered, the most productive registries make raw data available to the research community for further investigation
- Six of 7 ongoing registries have an operational website that provides contact information for potential patients. None of the closed registries have Web pages that describe the study (**Figure 6**)
- Thirteen registries resulted in at least 1 peer-reviewed publication

Figure 6. Accessibility of Data in Available Epidermolysis Bullosa Patient Registries Based on Presence of an (A) Operational Website and (B) Peer-Reviewed Publication

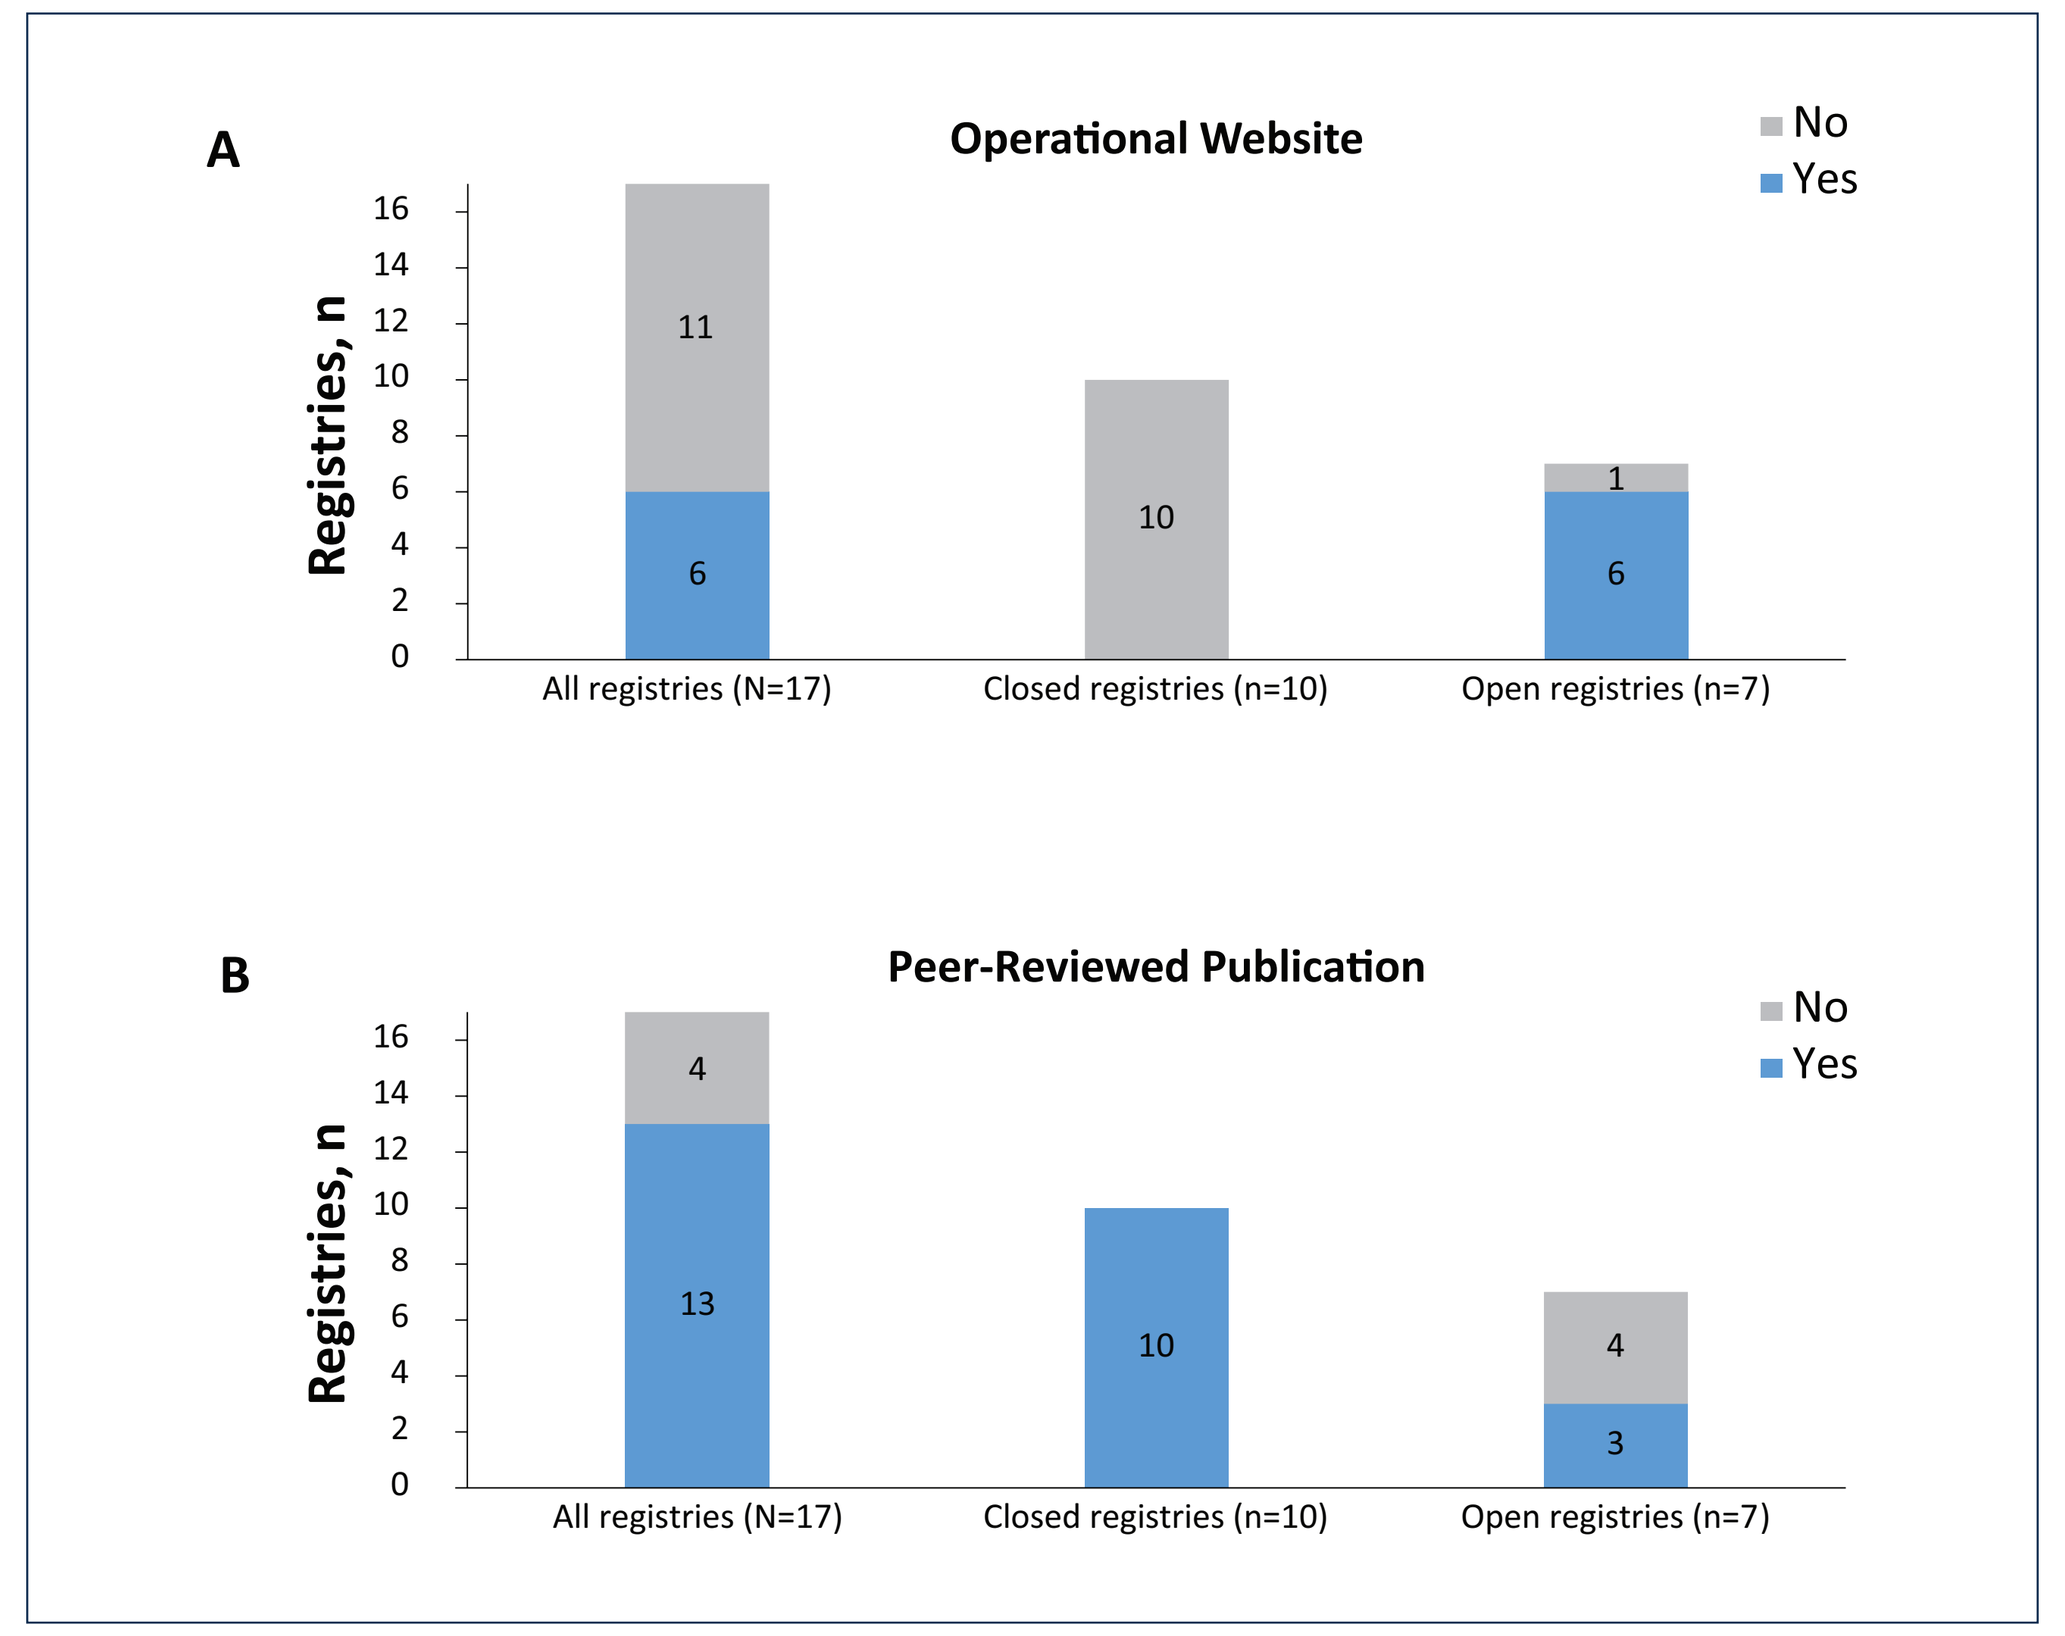
A
Operational Website
No
Yes
Registries, n
11
6
10
1
6
All registries (N=17)
Closed registries (n=10)
Open registries (n=7)
B
Peer-Reviewed Publication
No
Yes
Registries, n
4
13
10
4
3
All registries (N=17)
Closed registries (n=10)
Open registries (n=7)

OVERVIEW

Registry Characteristics

- An overview of the characteristics of each of the 17 unique epidermolysis bullosa registries identified are shown in **Table 1**

Table 1. Select Characteristics of Available Epidermolysis Bullosa Patient Registries (N = 17)

Registry	International (2+ countries)	>10-Year Duration	>500 Patients Enrolled	Proactive Search	Genotype Required	Includes Patient-Reported Outcomes	Ongoing	Publicly Accessible Raw Data	Operational Website	Resulted in a Peer-Reviewed Publication
1	■	■		■			■			■
2		■					■		■	
3			■				■		■	■
4		■								■
5						■	■		■	
6		■	■		■		■		■	
7	■		■		■		■	■	■	■
8	■			■	■					■
9		■	■	■						■
10			■							■
11		■		■						■
12		■								■
13										■
14				■						■
15										■
16						■	■		■	
17		■	■	■						■

Blue boxes indicate that the corresponding registry has the indicated characteristic. Gray boxes indicate that the registry does not have the indicated characteristic. Characteristics only reflect published and accessible data.

Registry Goals and Objectives

- Many registries stated multiple objectives when introducing the project, the most common being epidemiology (most commonly to determine simple incidence and prevalence data by subtype in a particular country or region) and natural history/clinical features (**Figure 7**)
- When comparing the goals of ongoing registries with the goals of those registries that have been discontinued, a shift from epidemiology to natural history/clinical features is evident
- The third most common objective, which was clearly stated in 2 registries, was the collection of patient information to facilitate recruitment in future clinical trials

Figure 7. Select Objectives of Available Epidermolysis Bullosa Patient Registries

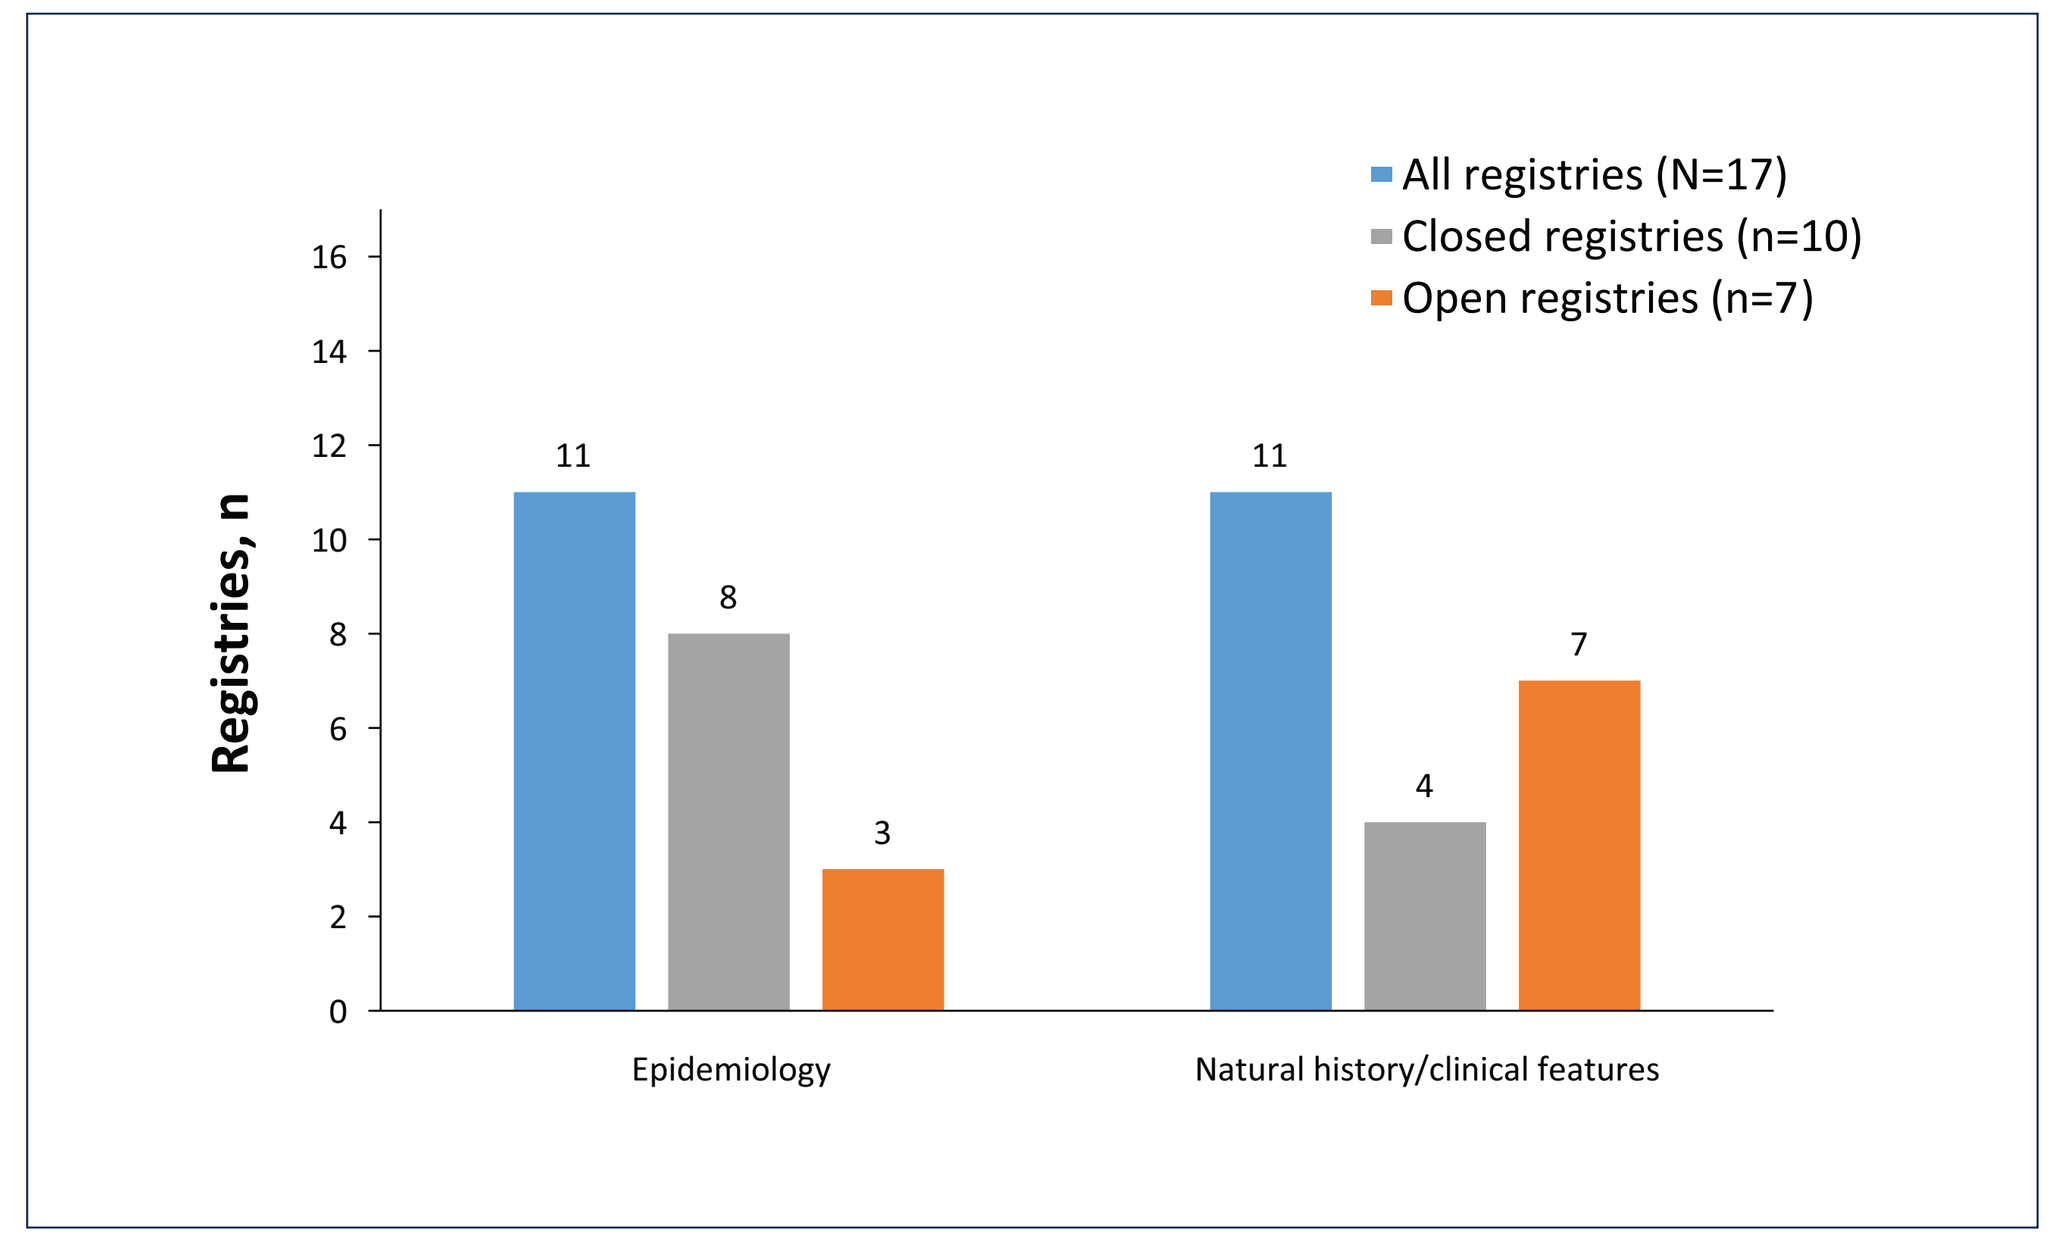
All registries (N=17)
Closed registries (n=10)
Open registries (n=7)
Registries, n
11
8
3
11
4
7
Epidemiology
Natural history/clinical features

One registry did not provide an objective. Some registries provided multiple objectives.

CONCLUSIONS AND RECOMMENDATIONS

Conclusions

- Despite the development of at least 17 disease registries, the understanding of epidermolysis bullosa is limited, particularly consensus on disease prevalence, genotype/phenotype relationships, and natural history
- A lack of early collaboration has resulted in the use of different registry methodologies, making comparisons between registries difficult
- Funding sources, geographic locations, inclusion criteria, types of data collected, and content varied across registries, leading to poor interoperability and lack of sustainability. Without international cooperation, these differences will prevent aggregation and comparison of data

Recommendations

- Future epidermolysis bullosa registries must adopt strategies used by other successful registries of rare diseases, such as standardized methodologies, a core common data set, funding and staffing to ensure sustainability, international collaborations to ensure registry interoperability, and policies that maximize data sharing
- To respect national rules, a governance group is necessary. That steering committee would oversee the identification of mandatory subsets of data that all registries would adhere to recording in their national registries
- Along with advances in technology, the understanding of epidermolysis bullosa continues to improve. Future epidermolysis bullosa registries must keep pace with evolving knowledge to contribute valuable disease information to the medical and patient communities

REFERENCES

- Coi A et al. *Public Health Genomics*. 2016;19(2):108-115.
- Irvine A et al. Epidermolysis Bullosa/European Reference Network Registry meeting summary; Dublin, Ireland; June 8, 2016. https://debraireland.org/wp-content/uploads/EB-registry-meeting-summary-Dublin-June-2016.pdf. Accessed June 21, 2017.

Registries: 1. Kho YC et al. *Arch Dermatol*. 2010;146(6):635-640. 2. Hintner H. The registry for epidermolysis bullosa (EB) in the EB House Austria: simple and effective. http://www.rare-diseases.eu/wp-content/uploads/2017/06/101.-A-registry-for-Epidermolysis-bullosa-EB-in-the-EB-House-Austria-Simple-and-Effective.pdf. Accessed June 21, 2017. 3. The Epidermolysis Bullosa Research Foundation. Research. http://www.ebmrf.org/research/. Accessed June 21, 2017. 4. Pavicić Z et al. *Pediatr Dermatol*. 1990;7:(2)108-110. 5. EB Care. Registry information sheet. https://ebcare.patientcrossroads.org/index.php?option=com_content&view=article&id=403&Itemid=479&lang=en. Accessed June 21, 2017. 6. Bruckner-Tuderman L. EB Center Freiburg. http://www.rare-diseases.eu/wp-content/uploads/2014/05/0101_Leena_BRUCKNER-TUDERMAN.pdf. Accessed June 21, 2017. 7. van den Akker PC et al. *Hum Mutat*. 2011;32(10):1100-1107. 8. Abu Sa'd J et al. *J Invest Dermatol*. 2006;126(4):777-781. 9. Tadini G et al. *G Ital Dermatol Venereol*. 2005;140(4):359-372. 10. Shinkuma S et al. *Dermatol Clin*. 2010;28(2):431-432, xvi. 11. McKenna KE et al. *Br J Dermatol*. 1992;127(4):318-321. 12. Yuen WY et al. *Br J Dermatol*. 2011;165(6):1314-1322. 13. Dănescu S et al. *J Eur Acad Dermatol Venereol*. 2015;29(5):899-903. 14. Horn HM et al. *Br J Dermatol*. 1997;136(4):560-564. 15. Davison BC. *Med Genet*. 1965;2(4):233-242. 16. Mellerio J. PEBLES: natural history of RDEB. http://www.debra-international.org/fileadmin/media_data/4_DEBRA_International/2_Events/2016_DI_Congress_Zagreb/PEBLES_-_Natural_history_of_RDEB_-_Jemima_Mellerio.pdf. Accessed June 21, 2017. 17. Fine JD. *JAMA Dermatol*. 2016;152(11):1231-1238.

ACKNOWLEDGMENTS

The authors acknowledge all patients with epidermolysis bullosa and their families who participate in epidermolysis bullosa registries to help advance knowledge of this disease. Third-party medical editorial assistance was provided by ApotheCom and was supported by Amicus Therapeutics, Inc.

DISCLOSURE

Conflicts of Interest

CB has served as an investigator for Amicus Therapeutics and received honoraria from Celgene, Expanscience, Sanofi, La Roche-Posay, and Novartis. AP has served as an investigator and consultant for Amicus Therapeutics. JW, JG, EC, and HL are employees of and own stock in Amicus Therapeutics.

Amicus Therapeutics

http://bit.ly/2958kHL

Supported by Amicus Therapeutics, Inc.

Presented at the 13th World Congress of Pediatric Dermatology; July 6-9, 2017; Chicago, IL, USA